ENCORE 601: A phase 2 study of entinostat in combination with pembrolizumab in patients with microsatellite stable metastatic colorectal cancer.

Nilofer Saba Azad[1], Keisuke Shirai[2], Autumn Jackson McRee[3], Mateusz Opyrchal[4], Douglas Buckner Johnson[5], Peter Ordentlich[6], Susan Brouwer[6], Serap Sankoh[6], Emmett V. Schmidt[7], Michael L. Meyers[8], Melissa Lynne Johnson[9]

[1]The Sidney Kimmel Comprehensive Cancer Center at Johns Hopkins University, Baltimore, MD; [2]Norris Cotton Cancer Center, Dartmouth-Hitchcock Medical Center, Lebanon, NH; [3]UNC Chapel Hill Lineberger Comprehensive Cancer Center, Chapel Hill, NC; [4]Roswell Park Comprehensive Cancer Center, Buffalo, NY; [5]Vanderbilt University Medical Center, Nashville, TN; [6]Syndax Pharmaceuticals, Inc., Waltham, MA; [7]Merck & Co., Inc., Kenilworth, NJ; [8]Syndax Pharmaceuticals, Inc., New York, NY; [9]Sarah Cannon Research Institute, Nashville, TN. For inquiries regarding this poster, please email: Nazad2@jhmi.edu

BACKGROUND

In studies reported to date, no objective responses to single agent PD-1 inhibitors have been observed in advanced/metastatic CRC (mCRC) patients with microsatellite stable (MSS)/mismatch repair proficient (pMMR) tumors[1]. Since most patients with mCRC have MSS/pMMR tumors, additional strategies are needed to improve effectiveness of immunotherapies in these patients[2].

Entinostat (ENT), an oral, class I-selective histone deacetylase inhibitor, enhances anti-PD-1 activity by downregulation of immunosuppressive cell types (eg, Tregs and MDSCs) in the tumor microenvironment in vivo[3] and has shown promising activity with pembrolizumab (PEMBRO) in patients with melanoma and lung cancer[4-6]. **Figure 1** describes the targets of entinostat and pembrolizumab in modulating immunosuppression.

This study evaluates the safety and efficacy of ENT + PEMBRO in patients with MSS/pMMR mCRC.

Figure 1. **Entinostat and Pembrolizumab Synergize to Modulate Immunosuppression.**

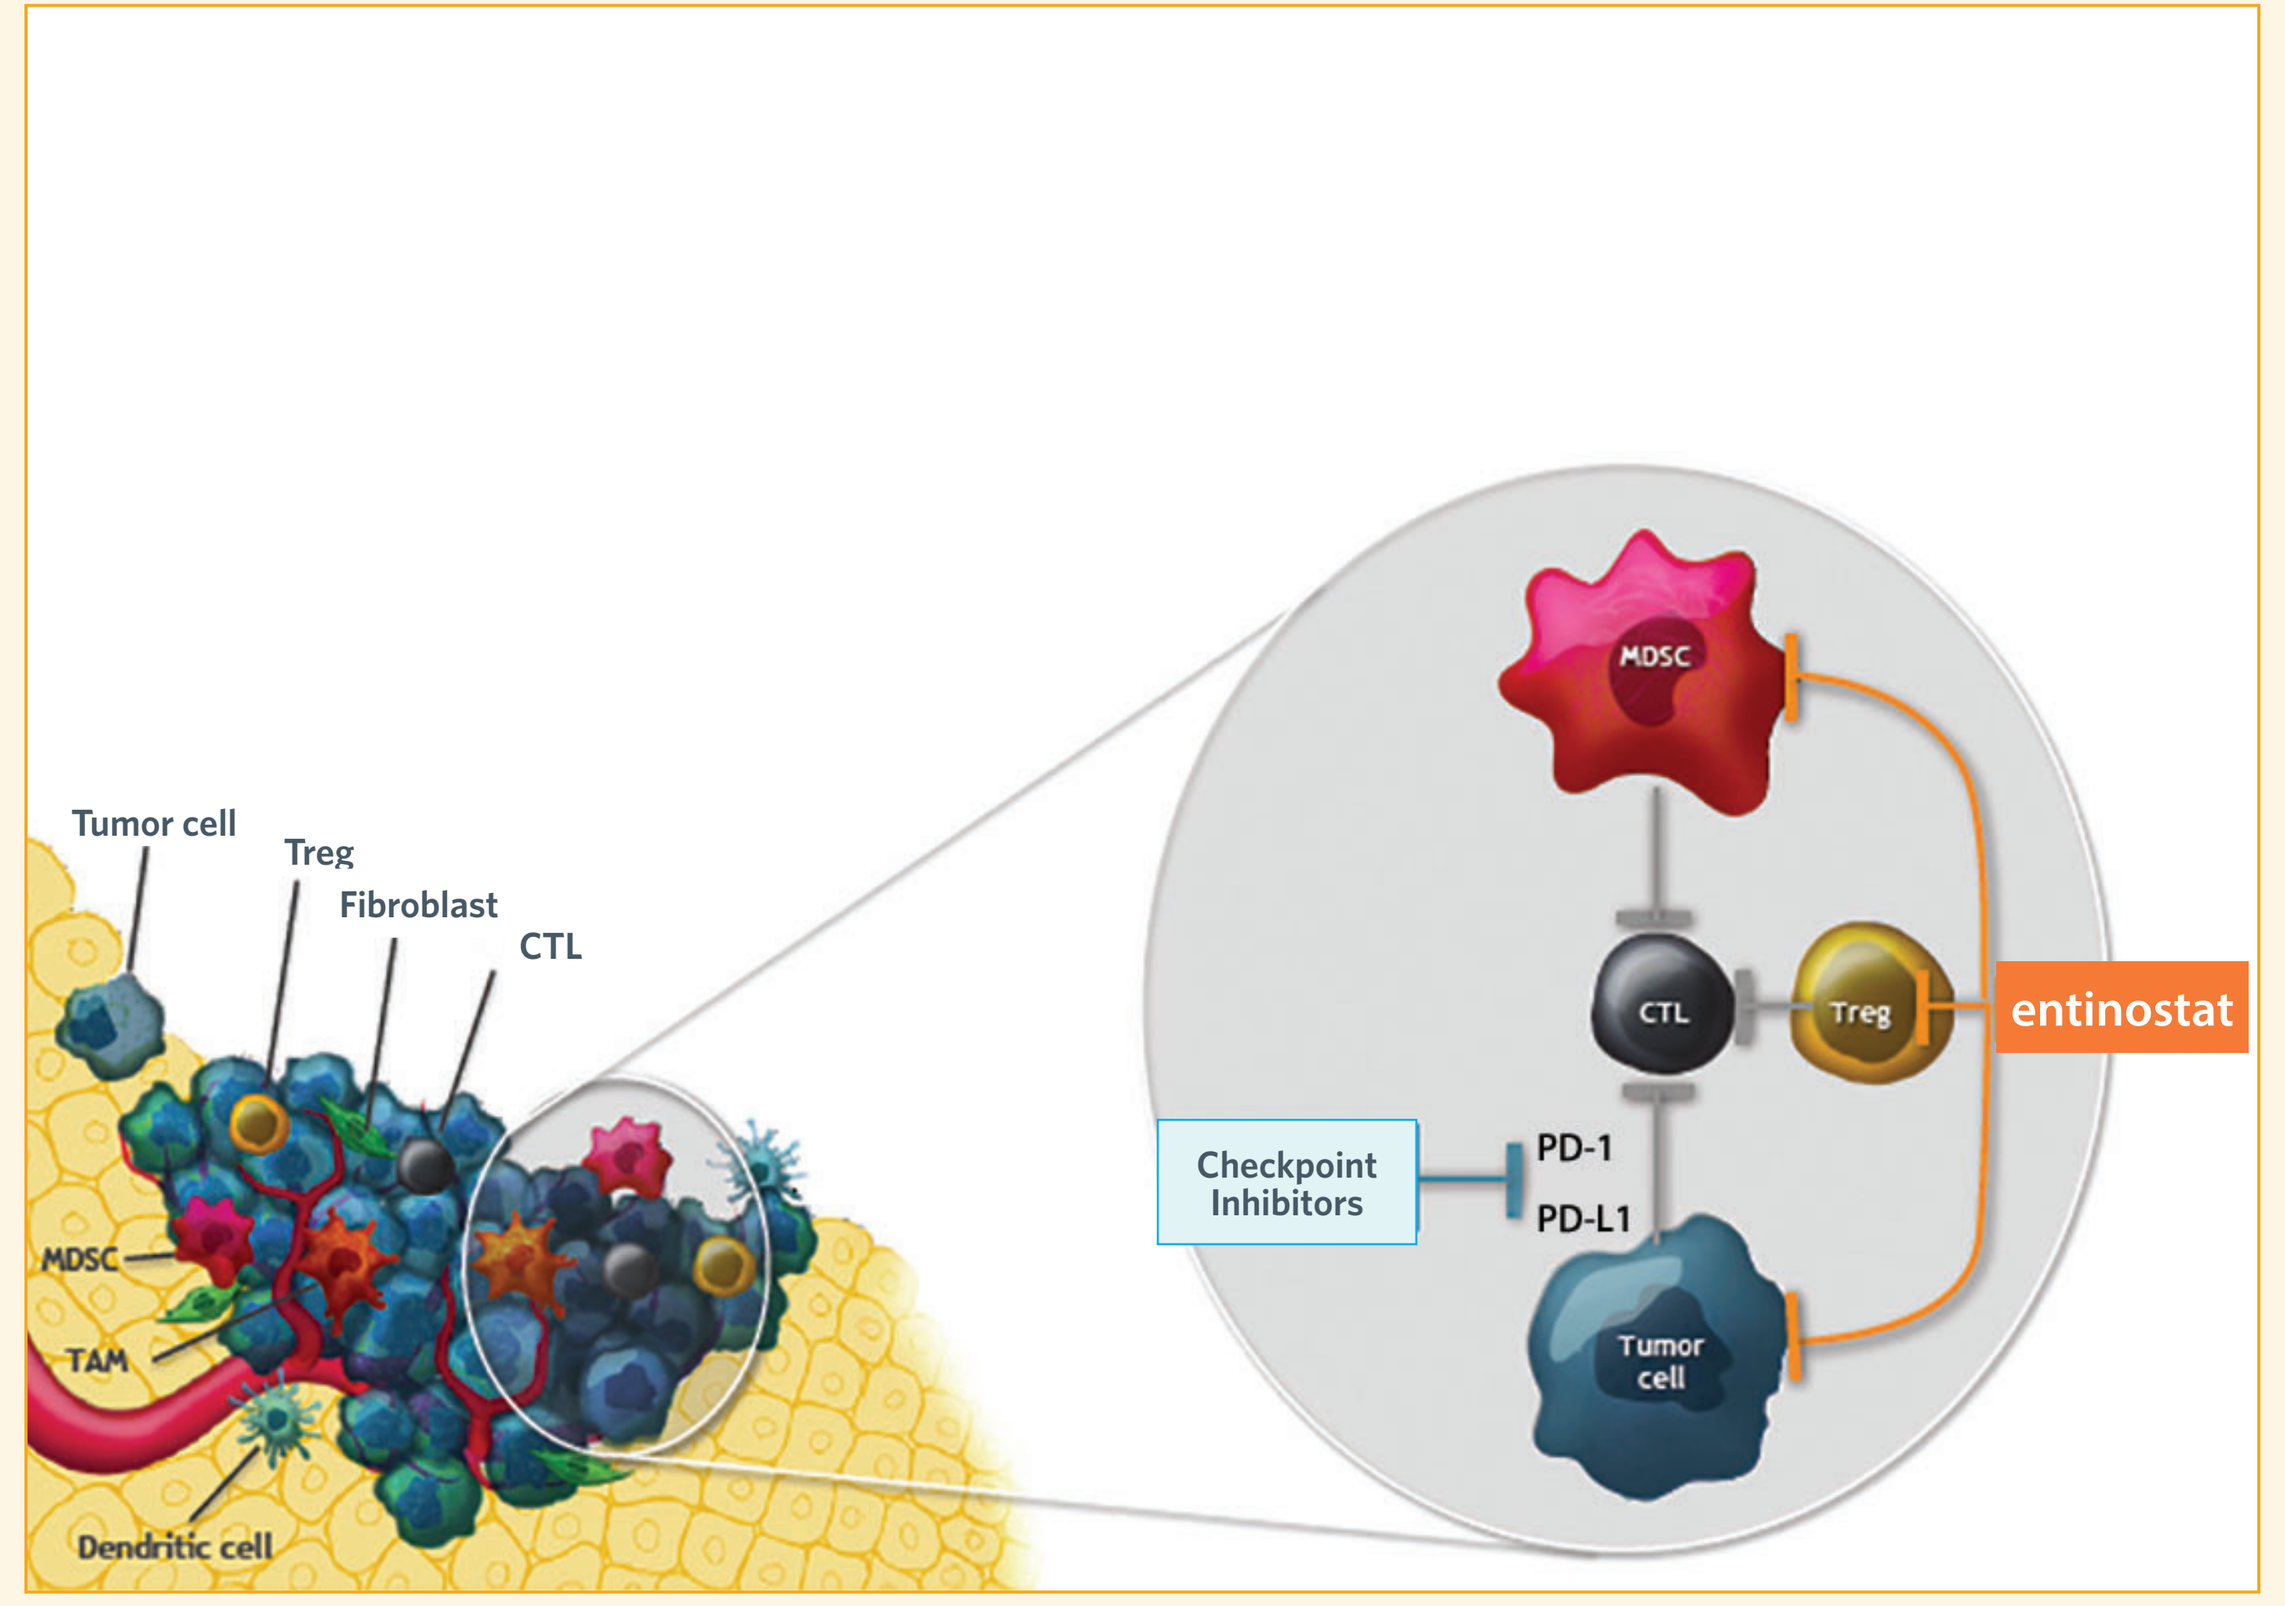
Tumor cell
Treg
Fibroblast
CTL
MDSC
TAM
Dendritic cell
MDSC
CTL
Treg
entinostat
Checkpoint Inhibitors
PD-1
PD-L1
Tumor cell

CTL, cytotoxic T lymphocyte; MDSC, myeloid derived suppressor cell; TAM, tumor-associated macrophage; Treg, regulatory T lymphocyte.

METHODS

Key Eligibility Criteria

- Documented mismatch repair-proficient colon cancer as determined by either immunohistochemistry for mismatch repair proteins or PCR-based functional microsatellite instability.
- Received at least 1 chemotherapeutic regimen in the advanced/metastatic setting and experienced documented, unequivocal progressive disease by either RECIST 1.1 or clinical assessment.
- Prior treatment with a PD-1/PD-L1-blocking antibody was not permitted.
- Other eligibility criteria included ECOG Performance Status 0 or 1, measurable disease per RECIST 1.1, and adequate end organ function.

METHODS (continued)

Study Design

- The Phase 2 expansion phase of ENCORE 601 utilizes a Simon 2-stage design to assess activity. The original design is noted below **(Figure 2)**, and the original Stage 1 patients reported here. The cutoff date for the analysis reported here was April 24, 2018.
- Based on emerging data regarding the minimal level of activity of PD-L1 inhibitors in MSS/pMMR CRC patients, the study was amended prior to completion of Stage 1 to expand Stage 1 enrollment to a total of 37 patients. Three responses of 37 are required to advance to Stage 2, at which point an additional 47 patients would be enrolled.
- The primary endpoint is objective response rate (ORR) as assessed by irRECIST assessed every 6 weeks.

Figure 2. **Simon 2-Stage Study Design.** The first 16 patients from stage 1 are reported

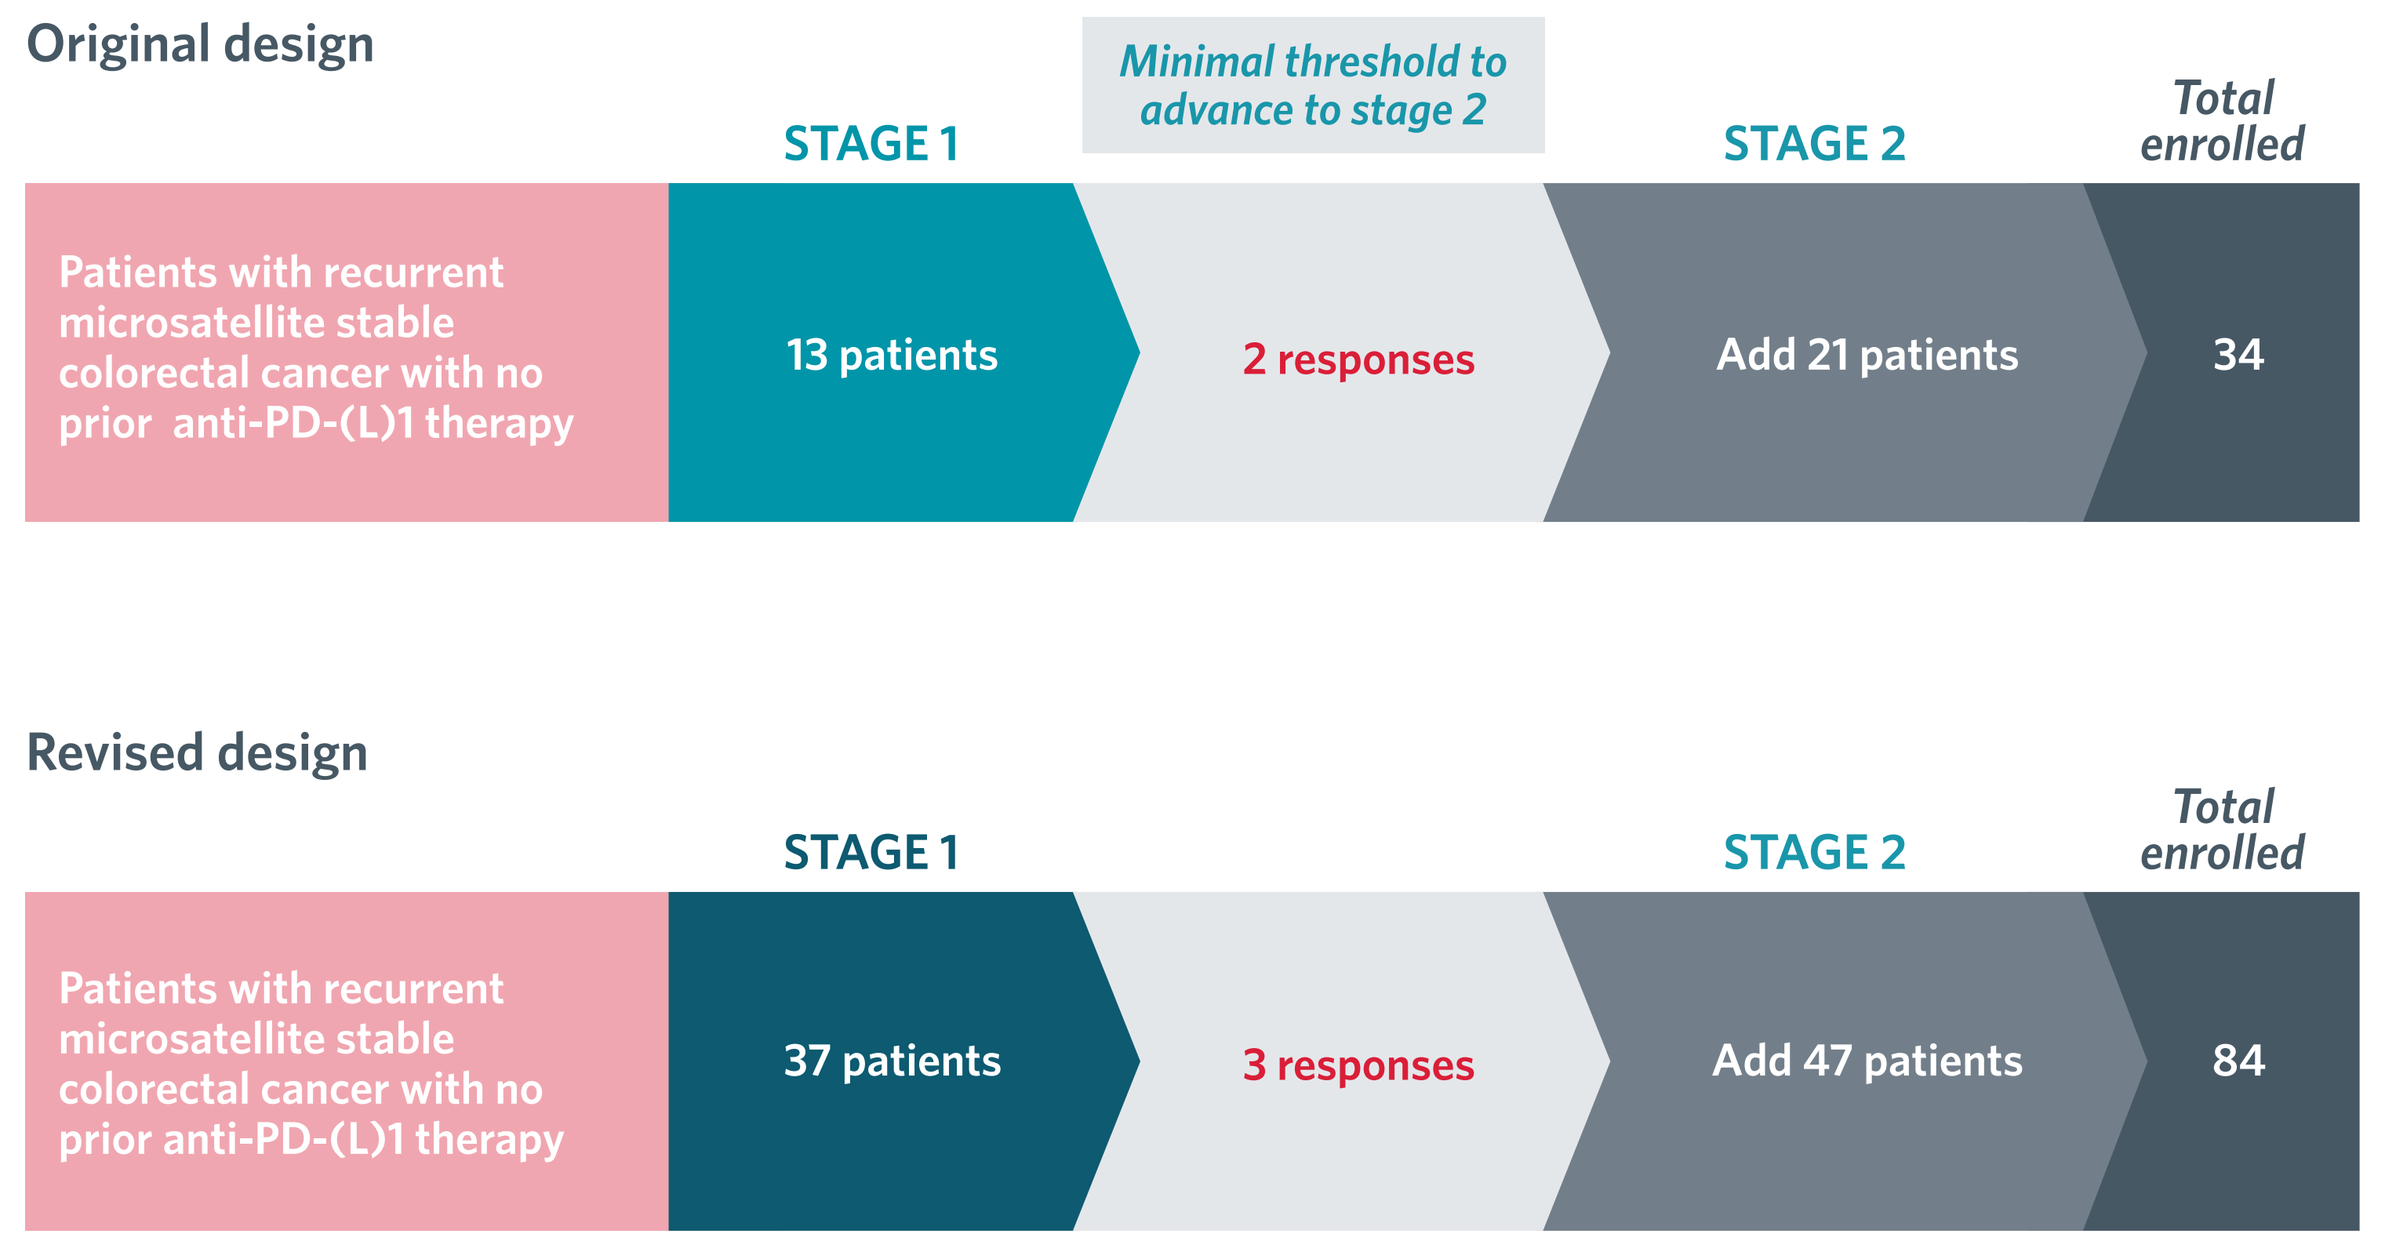
Original design
Minimal threshold to advance to stage 2
STAGE 1
STAGE 2
Total enrolled
Patients with recurrent microsatellite stable colorectal cancer with no prior anti-PD-(L)1 therapy
13 patients
2 responses
Add 21 patients
34
Revised design
STAGE 1
STAGE 2
Total enrolled
Patients with recurrent microsatellite stable colorectal cancer with no prior anti-PD-(L)1 therapy
37 patients
3 responses
Add 47 patients
84

Treatment Assessments

- Patients received entinostat 5 mg QW PO + pembrolizumab 200 mg Q3W IV in 21-day cycles until disease progression or discontinuation for other reasons.
- Peripheral blood and tumor tissue were collected for correlative studies are described in **Table 1**.

Table 1: Correlative Analysis	Blood Sample*	Tumor Tissue†
Protein lysine acetylation‡	Yes	Yes
MDSCs	Yes	Yes
T cells (CD8, CD4, Treg)	Yes	Yes
B cells	Yes	Yes
Dendritic cells	Yes	Yes
Natural killer cells	Yes	Yes
Macrophage	–	Yes
Gene expression - NanoString; RNA-Seq	–	Yes

*Samples are collected C1D1, C2D1, and C2D15.
†Samples are collected C1D1 and C2D15; analyses of markers prioritized based on tissue availability.
‡Samples are collected C1D1, C1D15, and C2D15.
CD, cluster of differentiation; C1D1, cycle 1, day 1; C1D15, cycle 1, day 15; C2D1, cycle 2, day 1; C2D15, cycle 2, day 15.

PATIENT DEMOGRAPHICS

- For the first 16 patients, the first patient enrolled August 2017 and last patient enrolled September 2017.
- Patient demographics are listed in **Table 2**.

Table 2: Demographics	Total (N=16)
Sex	
Male	8 (50%)
Female	8 (50%)
Age (years)	
Median	58.0
Range	36 to 69
Race	
White	14 (88%)
Black or African American	1 (6%)
Other	1 (6%)
Baseline ECOG Performance Score	
0	7 (44%)
1	8 (50%)
Stage of Disease at Initial Diagnosis	
IIA	1 (6%)
IIIA	3 (19%)
IIIB	2 (13%)
IVA	4 (25%)
IVB	5 (31%)
Baseline LDH (> ULN)	
Yes	5 (31%)
No	11 (69%)
Baseline CEA (> ULN)	
Yes	8 (50%)
No	2 (13%)
Not Reported	6 (38%)
Tumor Location at Initial Diagnosis	
Left side	2 (13%)
Right side	5 (31%)
Rectum	4 (25%)
Transverse	1 (6%)
Not specified	4 (25%)
Lines of Prior Therapy	
1	3 (19%)
2	4 (25%)
3	2 (13%)
4	4 (25%)
>4	3 (19%)

CEA, carcinoembryonic antigen; ECOG, Eastern Cooperative Oncology Group; LDH, lactate dehydrogenase; ULN, upper limit of normal.

EFFICACY

- One patient of the first 16 had an objective response, resulting in an ORR of 6% (95% CI: 0-30).
- The patient with a partial response is ongoing, as shown in **Figure 3** and **Figure 4**. Additional details of this patient are as follows:
 - 68 year old, male, with right sided primary, CEA at baseline = 9.8 ng/mL (ULN = 3.8 ng/mL), 6 lines of prior therapy in the advanced setting (best response to any of these prior therapies was stable disease), ECOG PS = 0, and sites of metastases = lung.
 - The patient entered the study 7.2 years following time of metastatic diagnosis (7.4 years from initial diagnosis).
 - MSS status confirmed by Foundation One.
 - Mutation burden = intermediate 10 muts/Mb
- Two patients (14-006 and 26-002) exhibited pseudoprogression at the first tumor assessment, as seen in **Figure 5**.
- Median time on treatment was 12.28 weeks, with the patient with a partial response being on treatment for over 30 weeks and ongoing.
- Nine patients experienced stable disease as best response **(Figure 3)**.
- Two of the nine experienced stable disease for 4 months or longer shown in **Figure 4**.
- Median progression-free survival was 86 days (95% CI, 46-134).

EFFICACY (continued)

Figure 3. **Percentage Change in Tumor Size from Baseline (Sum of Diameters of Target Lesions).** One partial response has been observed to date. Three patients did not have post-baseline measurements.

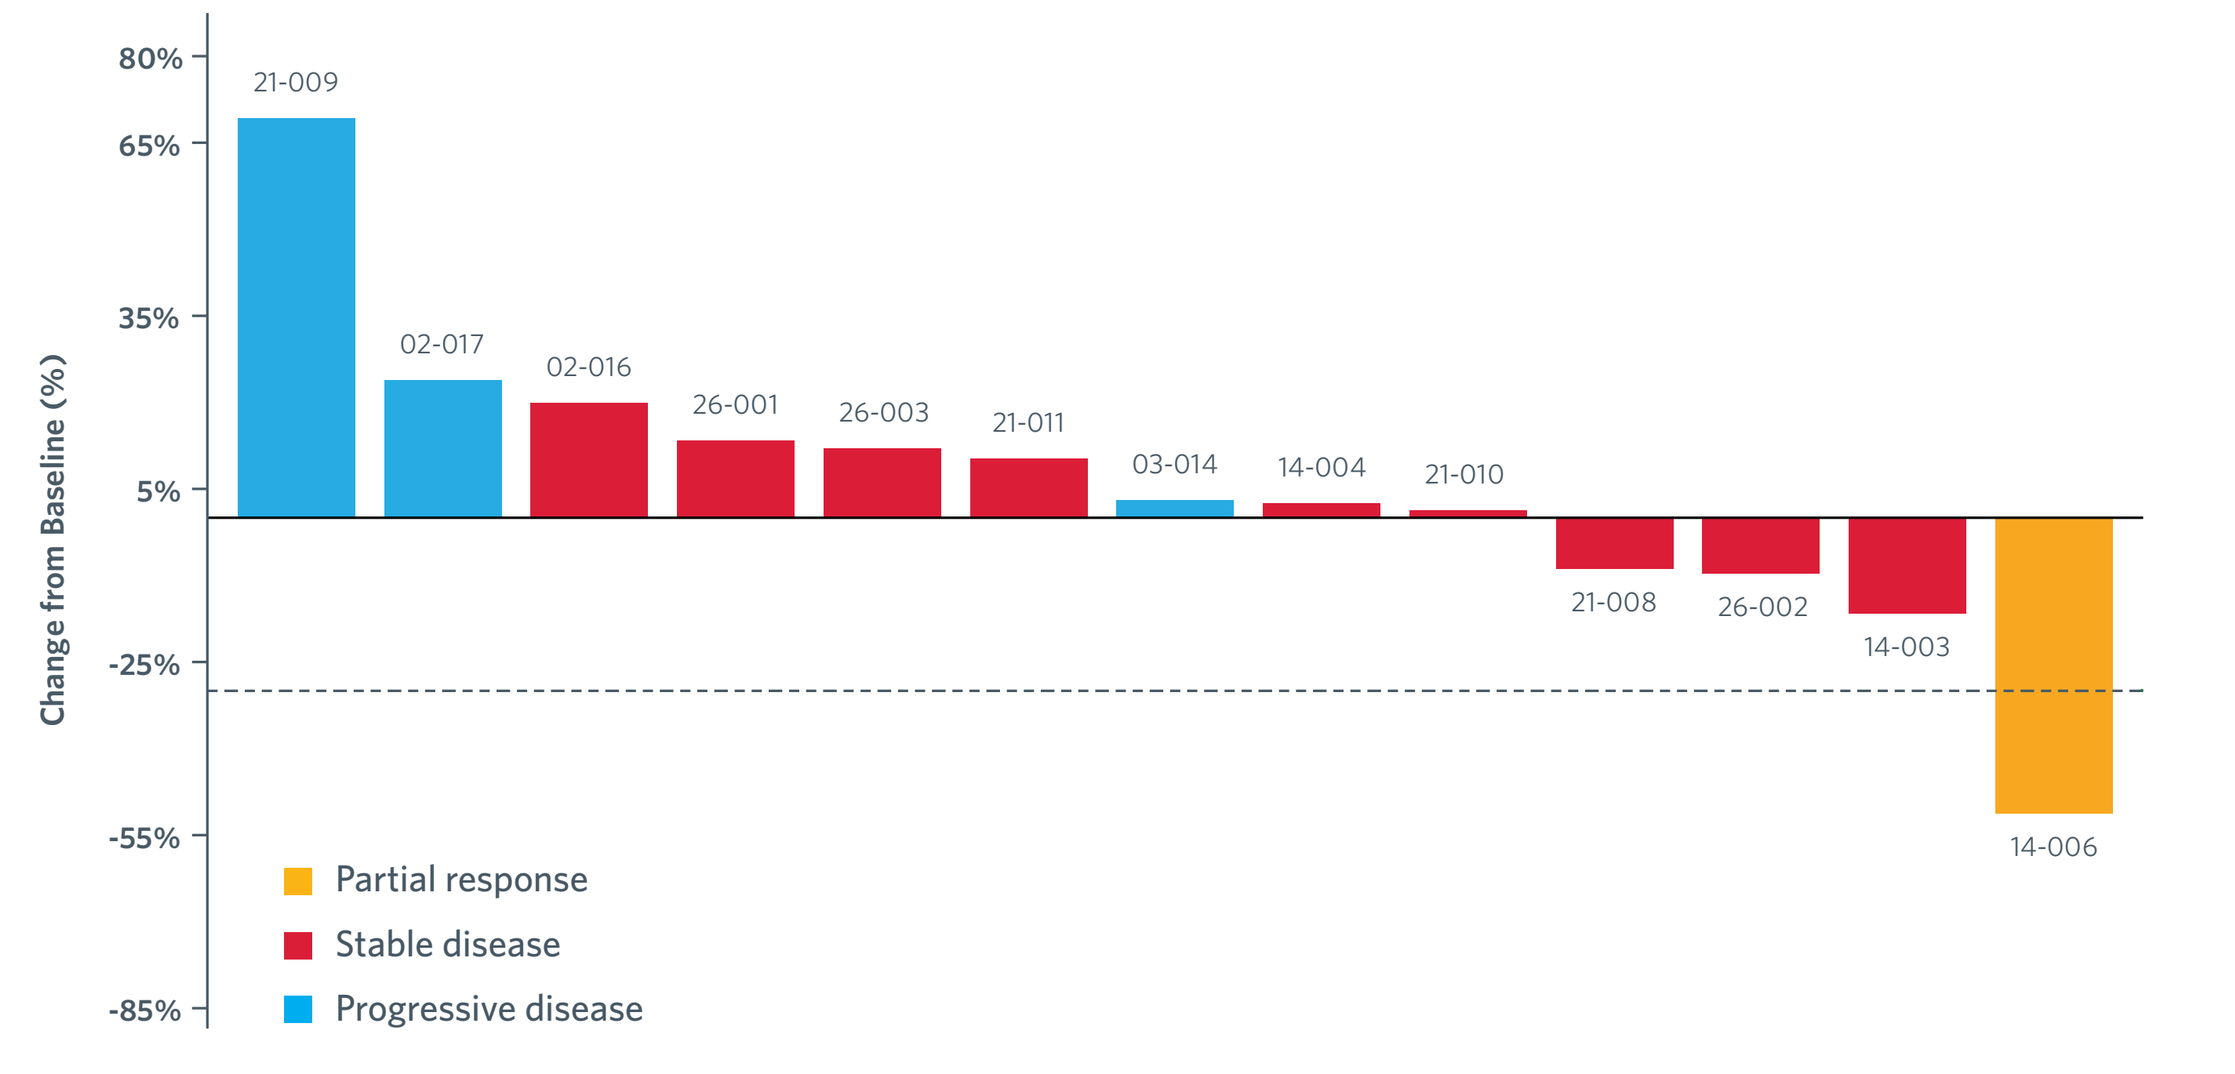
Change from Baseline (%)
80%
65%
35%
5%
-25%
-55%
-85%
21-009
02-017
02-016
26-001
26-003
21-011
03-014
14-004
21-010
21-008
26-002
14-003
14-006
Partial response
Stable disease
Progressive disease

Figure 4. **Patient Response and Time on Treatment.** irRECIST patient response by investigator assessment. Nine of 16 patients exhibited stable disease and one patient remains on treatment.

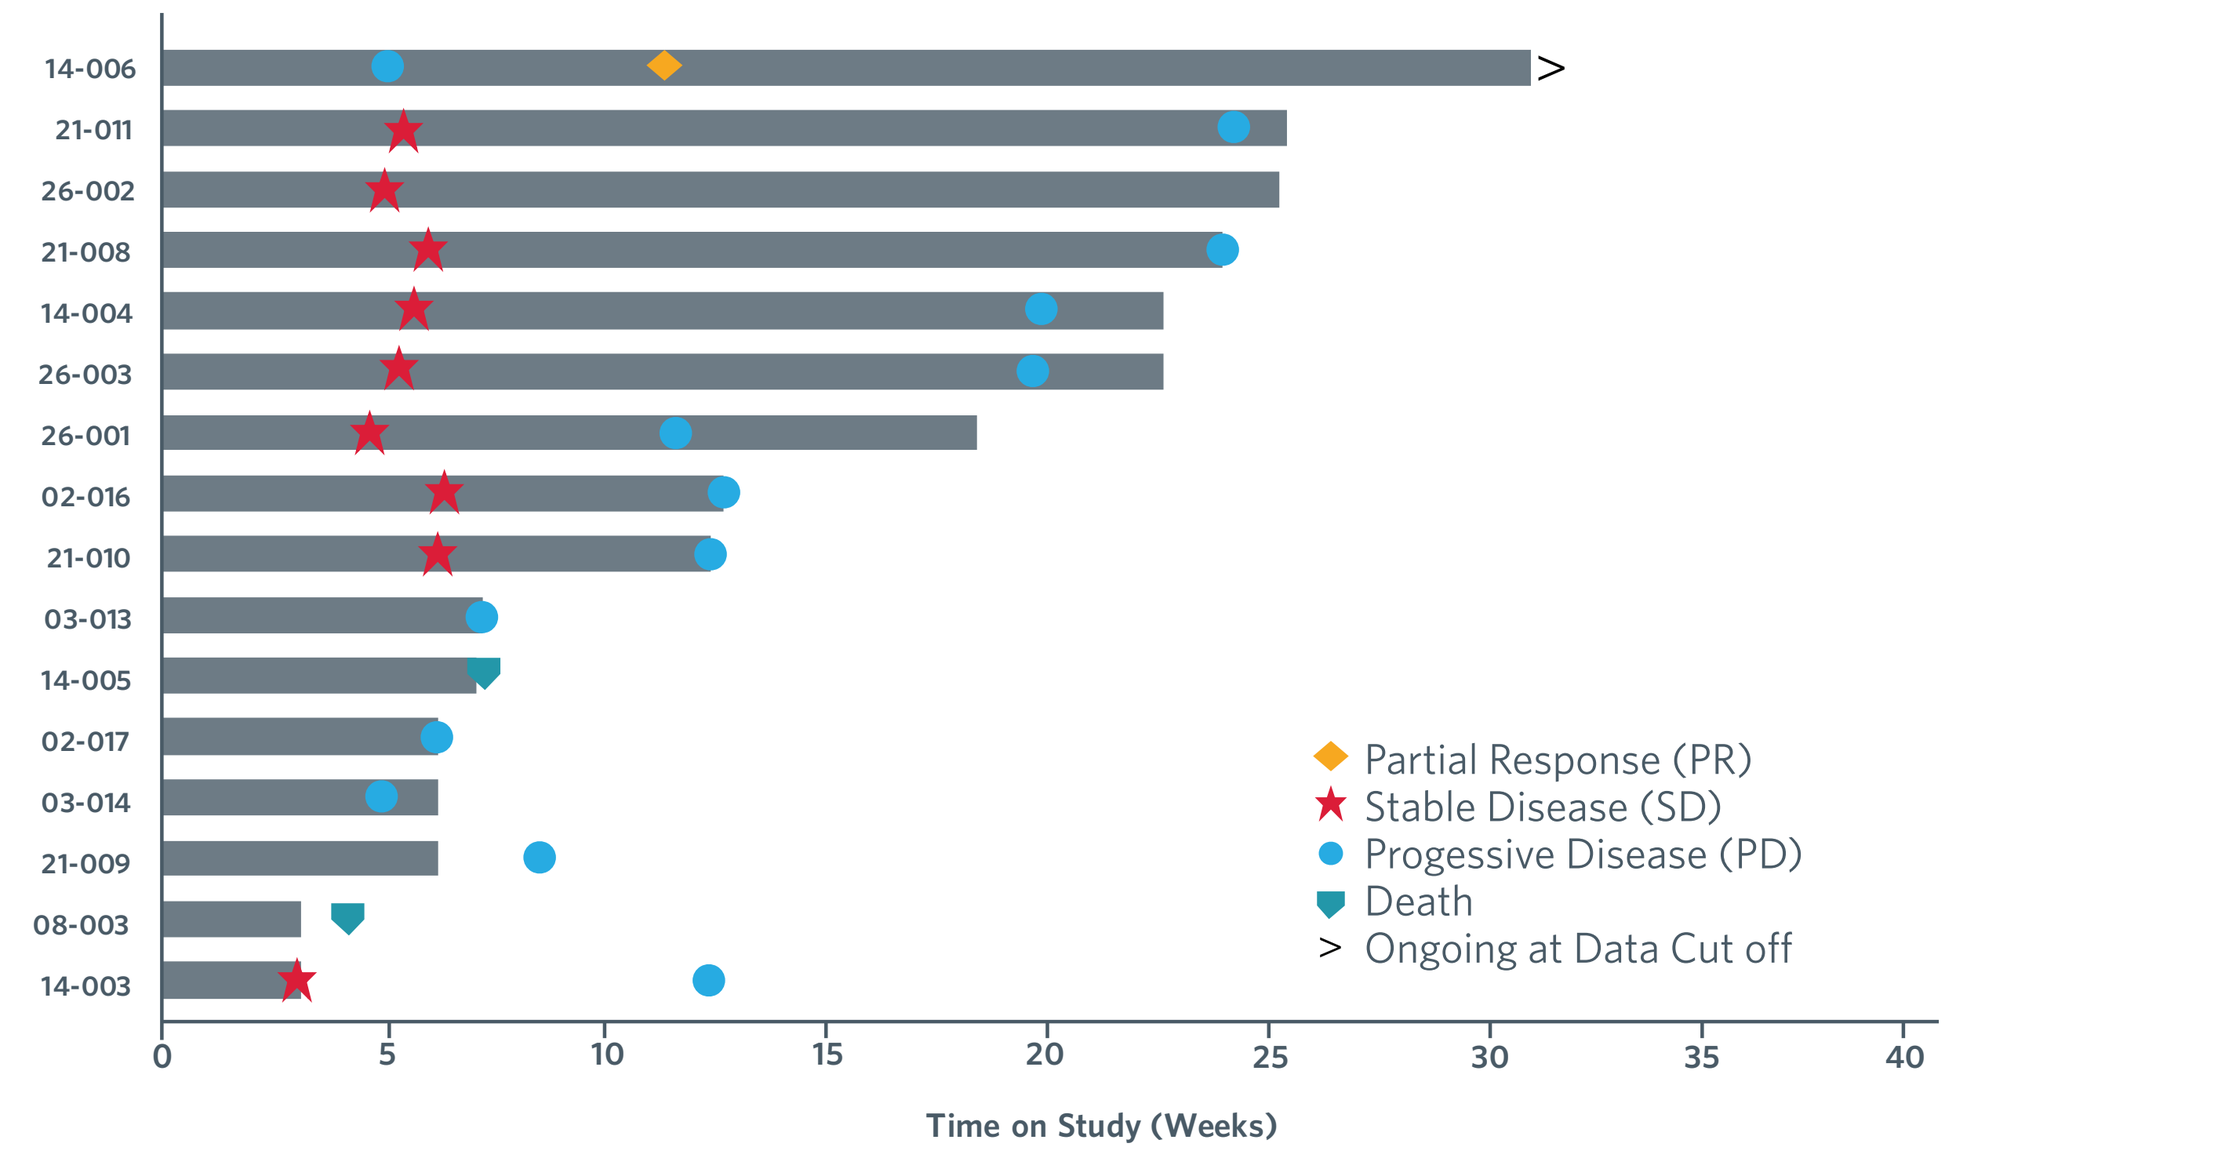
14-006
21-011
26-002
21-008
14-004
26-003
26-001
02-016
21-010
03-013
14-005
02-017
03-014
21-009
08-003
14-003
Partial Response (PR)
Stable Disease (SD)
Progessive Disease (PD)
Death
> Ongoing at Data Cut off
0
5
10
15
20
25
30
35
40
Time on Study (Weeks)

Figure 5. **Change in Tumor Size from Baseline Over Time.** Two patients exhibited pseudoprogression.

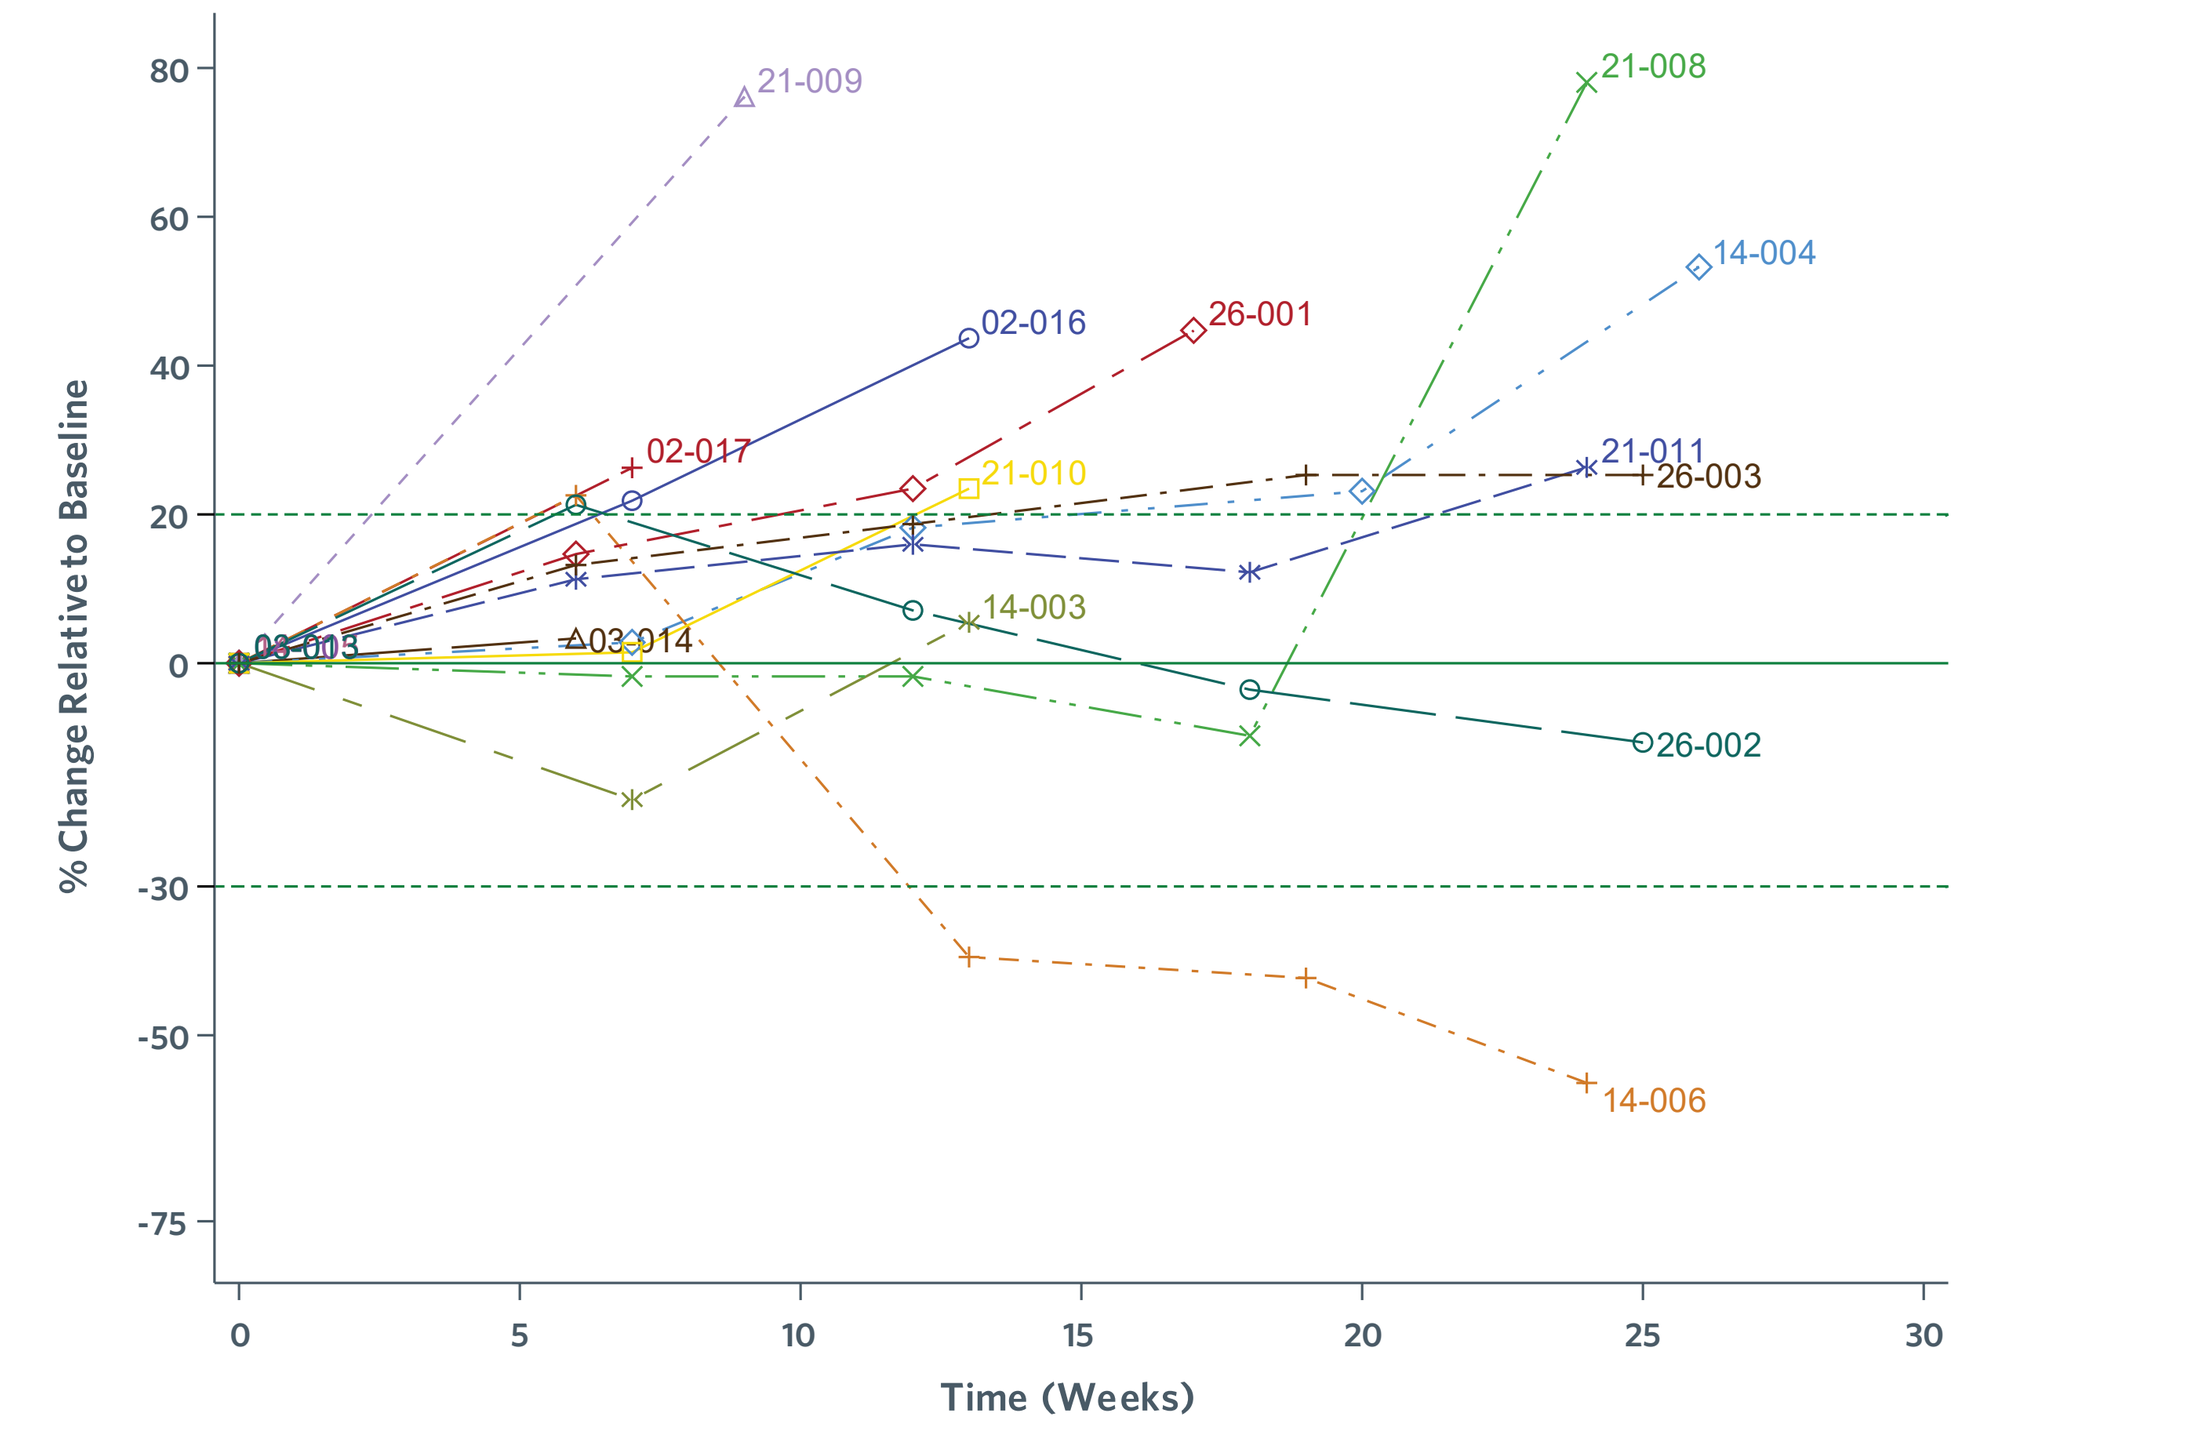
% Change Relative to Baseline
80
60
40
20
0
-30
-50
-75
21-009
21-008
14-004
02-016
26-001
02-017
21-010
21-011
26-003
14-003
03-003
03-014
26-002
14-006
0
5
10
15
20
25
30
Time (Weeks)

SAFETY

- Table 3 shows treatment-emergent adverse events related to entinostat or pembrolizumab or both in >10% of patients, or Grade ≥3 TEAE in any patient.
- TEAEs Grade 3 or higher occurred in 11/16 patients.
 - One patient experienced a Grade 3/4 related irAE (pneumonitis). Seven other patients experienced Grade 3/4 AEs with only anemia and blood alkaline phosphatase increase occurring in more than one patient (2 patients each).
- 2 (13%) patients discontinued due to a treatment-related AE. These events included liver enzyme increase and pneumonitis.

Table 3: Safety	Any Grade (N=16)	Grade ≥3 (N=16)
Subjects With At Least One Treatment-Emergent Adverse Event Related to Study Treatment	**16 (100.0)**	**8 (50.0)**
Gastrointestinal disorders		
Nausea	5 (31.3)	
Diarrhea	4 (25.0)	1 (6.3)
Vomiting	4 (25.0)	
Constipation	3 (18.8)	
Gastroesophageal reflux disease	2 (12.5)	
Pancreatitis	1 (6.3)	1 (6.3)
General disorders and administration site conditions		
Fatigue	8 (50.0)	1 (6.3)
Respiratory, thoracic and mediastinal disorders		
Pneumonitis	2 (12.5)	1 (6.3)
Skin and subcutaneous tissue disorders		
Pruritus	3 (18.8)	
Rash maculo-papular	2 (12.5)	
Investigations		
Blood alkaline phosphatase increased	6 (37.5)	2 (12.5)
Aspartate aminotransferase increased	5 (31.3)	1 (6.3)
Platelet count decreased	3 (18.8)	
Lymphocyte count decreased	1 (6.3)	1 (6.3)
Neutrophil count decreased	1 (6.3)	1 6.3)
White blood cell count decreased	1 (6.3)	1 (6.3)
Metabolism and nutrition disorders		
Decreased appetite	3 (18.8)	
Hypoalbuminemia	2 (12.5)	
Hyperglycemia	1 (6.3)	1 (6.3)
Hypophosphatemia	1 (6.3)	1 (6.3)
Musculoskeletal and connective tissue disorders		
Arthralgia	4 (25.0)	
Myalgia	4 (25.0)	
Endocrine disorder		
Hypothyroidism	2 (12.5)	
Blood and lymphatic system disorders		
Anemia	4 (25.0)	2 (12.5)
Thrombocytopenia	2 (12.5)	
Nervous system disorder		
Headache	2 (12.5)	
Hepatobiliary disorder		
Cholangitis	1 (6.3)	1 (6.3)

CONCLUSIONS

- ENT + PEMBRO demonstrates acceptable safety and encouraging preliminary activity in a small cohort of MSS/pMMR CRC patients, a patient population in which clinical benefit has not been observed with PD-(L)1 monotherapy.
- Additional analyses are ongoing to identify biomarkers that can predict clinical benefit in this population.
- The decision to proceed to Stage 2 will be based on the analyses of the amended Stage 1 cohort. Enrollment is currently ongoing.

References:

1. Kather JN, et al. *Semin Cancer Biol.* 2018 [Epub Ahead of Print].
2. Cancer Genome Atlas Network. *Nature.* 2012;487:330-337.
3. Orillion A, et al. *Clin Cancer Res.* 2017;23:5187-5201.
4. Johnson ML, et al. Presented at the ASCO Annual Meeting, 2017. Abstract 9529.
5. Johnson ML, et al. Presented at SITC Annual Meeting, 2017. Abstract P72.
6. Gandhi L, et al. Presented at SITC Annual Meeting, 2017. Abstract O19.

Acknowledgments

The authors thank the patients and their families, investigators and study staff. This study was sponsored by Syndax Pharmaceuticals, Inc., in collaboration with Merck & Co., Inc., Kenilworth, NJ. Writing and editorial assistance was provided by BluPrint Oncology, LLC. This assistance was funded by Syndax Pharmaceuticals, Inc.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® and the author of this poster.

Poster presented at the American Society of Clinical Oncology Annual Meeting, June 1-5, 2018, Chicago IL.