Efficacy and safety of entinostat (ENT) and pembrolizumab (PEMBRO) in patients with melanoma progressing on or after a PD-1/L1 blocking antibody.

Sanjiv S. Agarwala[1], Stergios J. Moschos[2], Melissa Lynne Johnson[3], Mateusz Opyrchal[4], Dmitry Gabrilovich[5], Patrick Danaher[6], Fang Wang[5], Susan Brouwer[6], Peter Ordentlich[6], Serap Sankoh[6], Emmett V. Schmidt[7], Michael L. Meyers[8], Ryan J. Sullivan[9]

[1]St. Luke's Hospital, Easton, PA; [2]University of North Carolina Lineberger Comprehensive Cancer Center, Chapel Hill, NC; [3]Sarah Cannon Research Institute, Nashville, TN; [4]Roswell Park Comprehensive Cancer Center, Buffalo, NY; [5]The Wistar Institute, Philadelphia, PA; [6]NanoString Technologies, Inc., Seattle, WA; [6]Syndax Pharmaceuticals, Inc., Waltham, MA; [7]Merck & Co., Inc., Kenilworth, NJ; [8]Syndax Pharmaceuticals, Inc., New York, NY; [9]Massachusetts General Hospital, Boston, MA. For inquiries regarding this poster, please email: ssagarwala0501@gmail.com

BACKGROUND

Immune checkpoint inhibitors (ICIs) have improved prognosis for patients with advanced melanoma, but there is an unmet need for patients who progress after ICI[1].

Entinostat (ENT), an oral, class I-selective histone deacetylase inhibitor, enhances anti-PD-1 activity by downregulation of immunosuppressive cell types in the tumor microenvironment in vivo[2] **(Figure 1)** and has shown promising activity with pembrolizumab (PEMBRO) in patients with melanoma and lung cancer[3-5]. **Figure 1** describes the targets of entinostat and pembrolizumab in modulating immunosuppression.

Figure 1. **Entinostat and Pembrolizumab Synergize to Modulate Immunosuppression.**

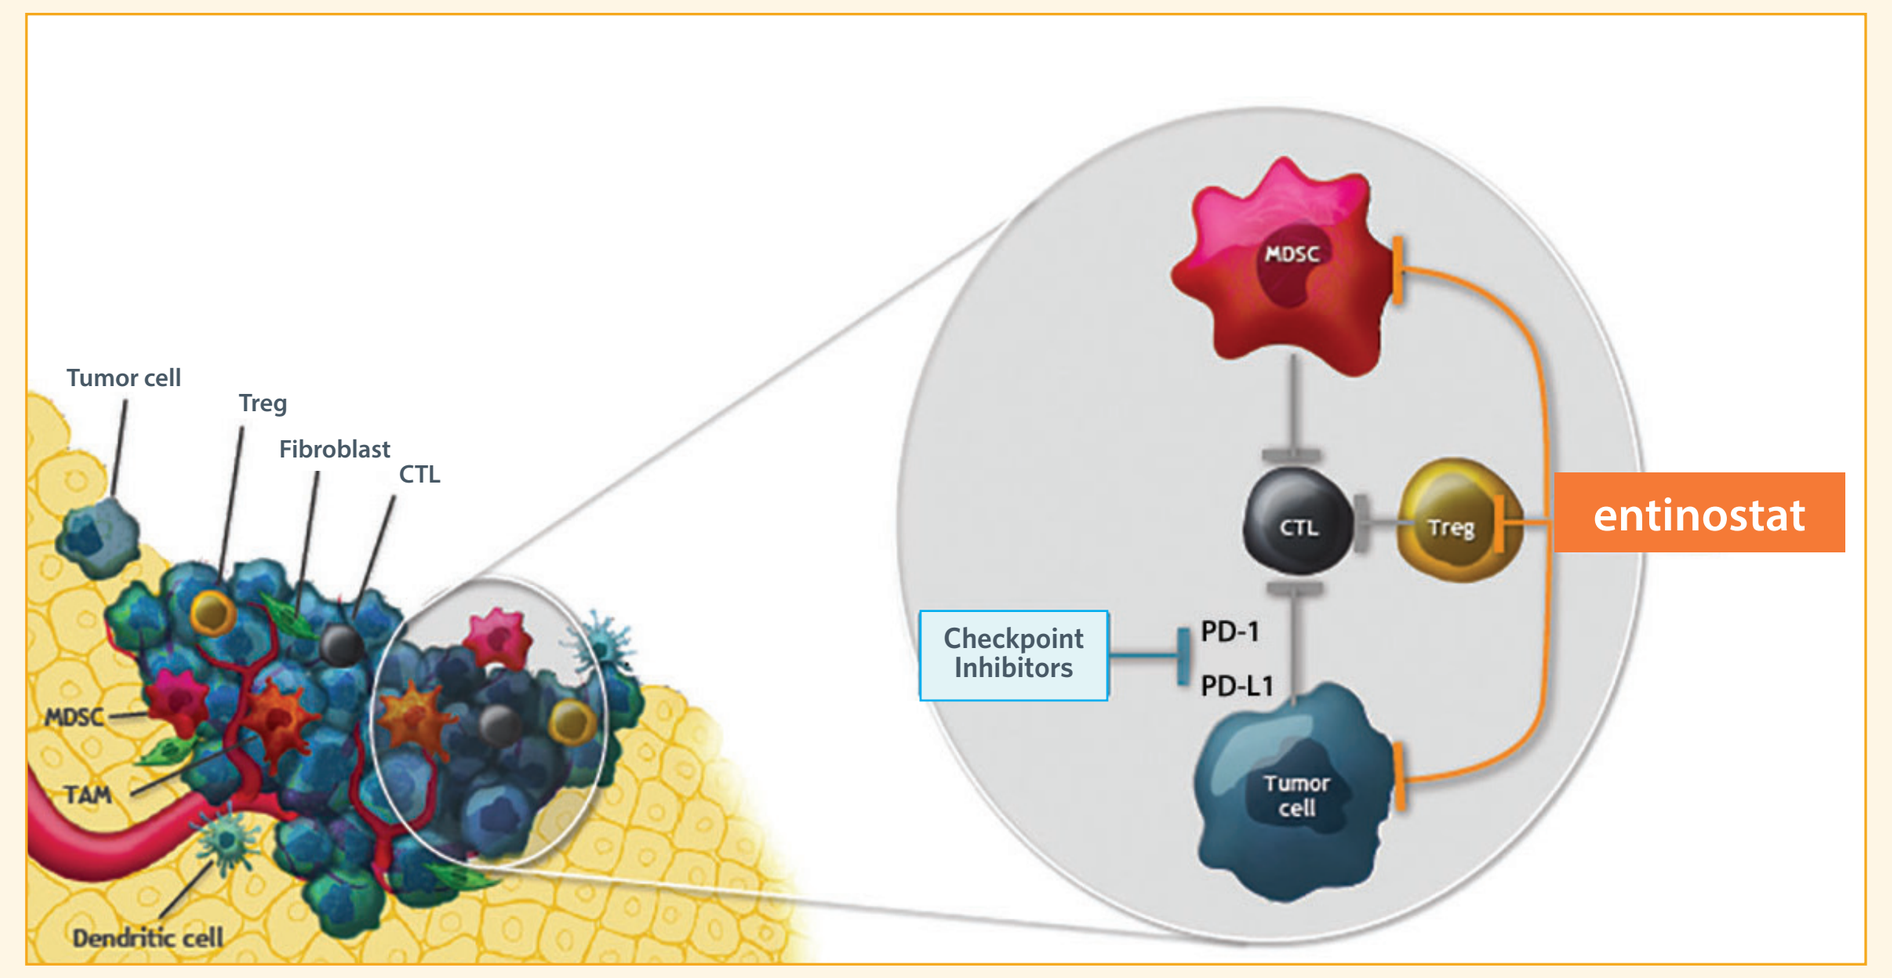
Tumor cell
Treg
Fibroblast
CTL
MDSC
TAM
Dendritic cell
MDSC
CTL
Treg
entinostat
Checkpoint Inhibitors
PD-1
PD-L1
Tumor cell

CTL, cytotoxic T lymphocyte; MDSC, myeloid derived suppressor cell; TAM, tumor-associated macrophage; Treg, regulatory T lymphocyte.

METHODS

Key Eligibility Criteria

- Histologically- or cytologically-confirmed diagnosis of unresectable or metastatic melanoma and previously treated with and unequivocally progressed on either a PD-1 or PD-L1-blocking antibody.
- If BRAF V600 mutation-positive, unequivocally progressed during treatment with a BRAF inhibitor.
- Other eligibility criteria included ECOG Performance Status 0 or 1, measurable disease per RECIST 1.1, and willingness to have fresh tumor samples collected during screening and at other time points as necessary.

Study Design

- The Phase 2 expansion phase of ENCORE 601 utilizes a Simon 2-stage design to assess activity. Four responses in 13 patients were observed in Stage 1 of the study, meeting the criteria to expand to Stage 2 and enroll up to 34 patients[3]. Data from the original 34 patients are reported here. The study was further revised to accrue up to 52 patients to increase statistical power and decrease Type I error.
- The cutoff date for the analysis reported here was April 24, 2018.
- The primary endpoint is overall response rate (ORR) as assessed by irRECIST.

Treatment Assessments

- Patients received entinostat 5 mg QW PO + pembrolizumab 200 mg Q3W IV in 21-day cycles until disease progression or discontinuation for other reasons.
- Peripheral blood and tumor tissue collected for correlative studies are described in **Table 1**.

METHODS (continued)

Table 1: Correlative Analysis	Blood Sample*	Tumor Tissue†
Protein lysine acetylation‡	Yes	Yes
MDSCs	Yes	Yes
T cells (CD8, CD4, Treg)	Yes	Yes
B cells	Yes	Yes
Dendritic cells	Yes	Yes
Natural killer cells	Yes	Yes
PD-L1	–	Yes
Macrophage	–	Yes
Gene expression - NanoString; RNA-Seq	–	Yes

*Samples are collected C1D1, C2D1, and C2D15.
†Samples are collected C1D1 and C2D15; analyses of markers prioritized based on tissue availability.
‡Samples are collected C1D1, C1D15, and C2D15.
CD, cluster of differentiation; C1D1, cycle 1, day 1; C1D15, cycle 1, day 15; C2D1, cycle 2, day 1; C2D15, cycle 2, day 15.

PATIENT DEMOGRAPHICS

Demographics

- The first patient was enrolled August 2016, and last patient was enrolled July 2017.
- Of the first 34 patients enrolled, 16 had refractory disease to prior PD-(L)1 therapy, and only 2 had a documented prior response.
- Median duration of prior PD-(L)1 therapy was < 6 months and the median time from last dose was 64 days.
- Twenty-two patients had prior ipilimumab & PD-(L)1 therapy and 7 had prior exposure to a BRAF inhibitor.
- Additional baseline demographics are listed in **Table 2.**

Table 2: Demographics	Total N=34
Sex	
Male	22 (65%)
Female	12 (35%)
Age (years)	
Median	64.0
Range	20 to 86
Race	
White	32 (94%)
Black or African American	1 (3%)
Other	1 (3%)
ECOG Performance Score	
0	19 (56%)
1	15 (44%)
PD-L1 Expression	
Negative	9 (26%)
Positive	17
Not Evaluable	8 (24%)
Visceral Metastases	
Yes	15 (44%)
No	17 (50%)
Missing	2 (6%)
LDH (> ULN)	
Yes	13 (38%)
No	21 (62%)
Previously Treatead with Ipilimumab & PD-1	
Yes	22 (65%)
No	12 (35%)
Previously Treated with BRAF Inhibitor	
Yes	7 (21%)
No	27 (79%)
Best Response on prior PD-1/PD-L1 Therapy	
Complete Response	1 (3%)
Partial Response	1 (3%)
Stable Disease	13 (38%)
Disease Progression	16 (47%)
Not Available	3 (9%)
Duration on Prior PD-1/PD-L1 Therapy (Days)	
Median	162.0
Range	22 to 702
Duration between last dose of prior PD-1/PD-L1 Therapy and First of dose of Study Therapy (Days)	
Median	64.0
Range	22 to 1128

ECOG, Eastern Cooperative Oncology Group; LDH, lactic acid dehydrogenase; PD-1, programmed cell death-1; PD-L1; programmed death ligand-1; ULN, upper limit of normal.

EFFICACY

- Partial response was observed in 9/34 patients as shown in **Figure 2**.
 - 6 partial responses were confirmed, resulting in an ORR of 18% (95% CI: 6.8-34.5)
- The longest response is over 80 weeks, with many still ongoing **(Figure 3)**.
- Stable disease was best response in 15/34 (44%) patients **(Figure 2)**.
- The median duration of response was 9.0 months (range: 2.1-14.4) **(Figure 4)**.
- Median progression-free survival was 90 days.
- Clinical benefit rate, defined as complete or partial response or stable disease for 24 weeks or more from initiation of treatment, was 29% (10/34 patients).
- At the data cutoff, four patients remain on treatment.

Figure 2. **Percentage Change in Tumor Size from Baseline (Sum of Diameters of Target Lesions).** Nine partial responses have been observed to date.

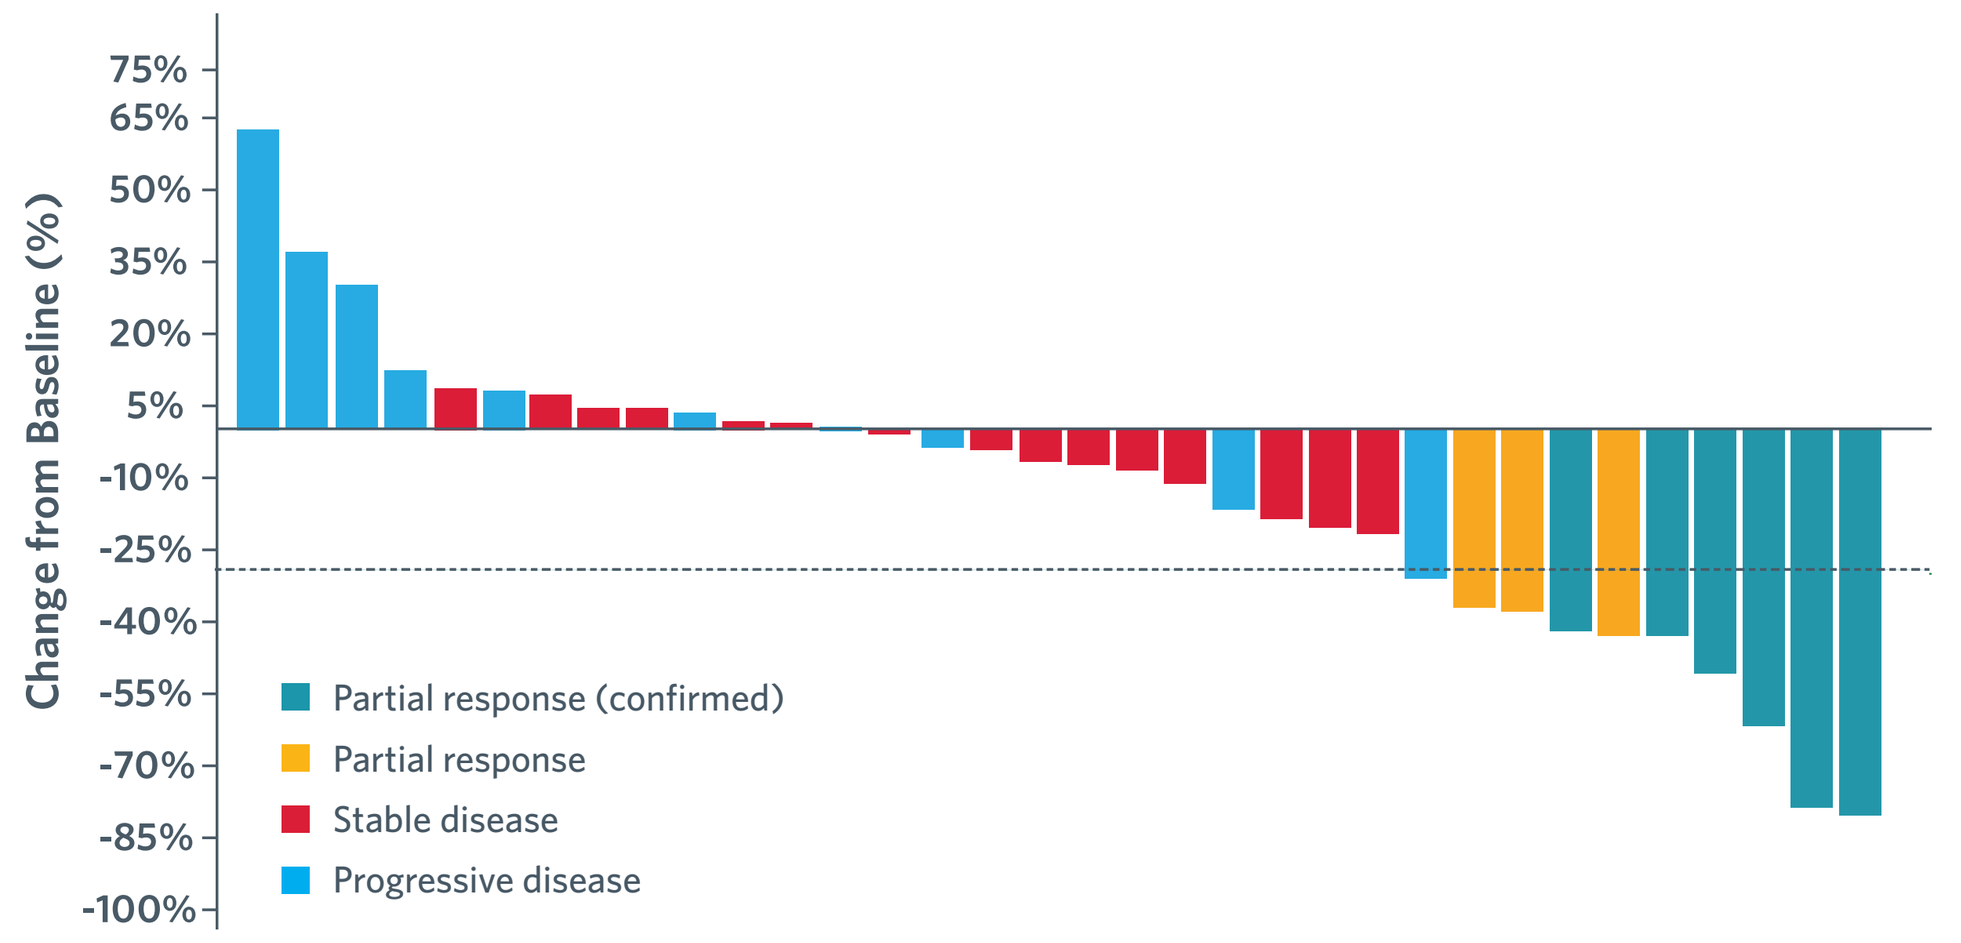
75%
65%
50%
35%
20%
5%
-10%
-25%
-40%
-55%
-70%
-85%
-100%
Change from Baseline (%)
Partial response (confirmed)
Partial response
Stable disease
Progressive disease

Figure 3. **Patient Response and Time on Treatment. irRECIST patient response by investigator assessment.** Three patients have received treatment for >1 year.

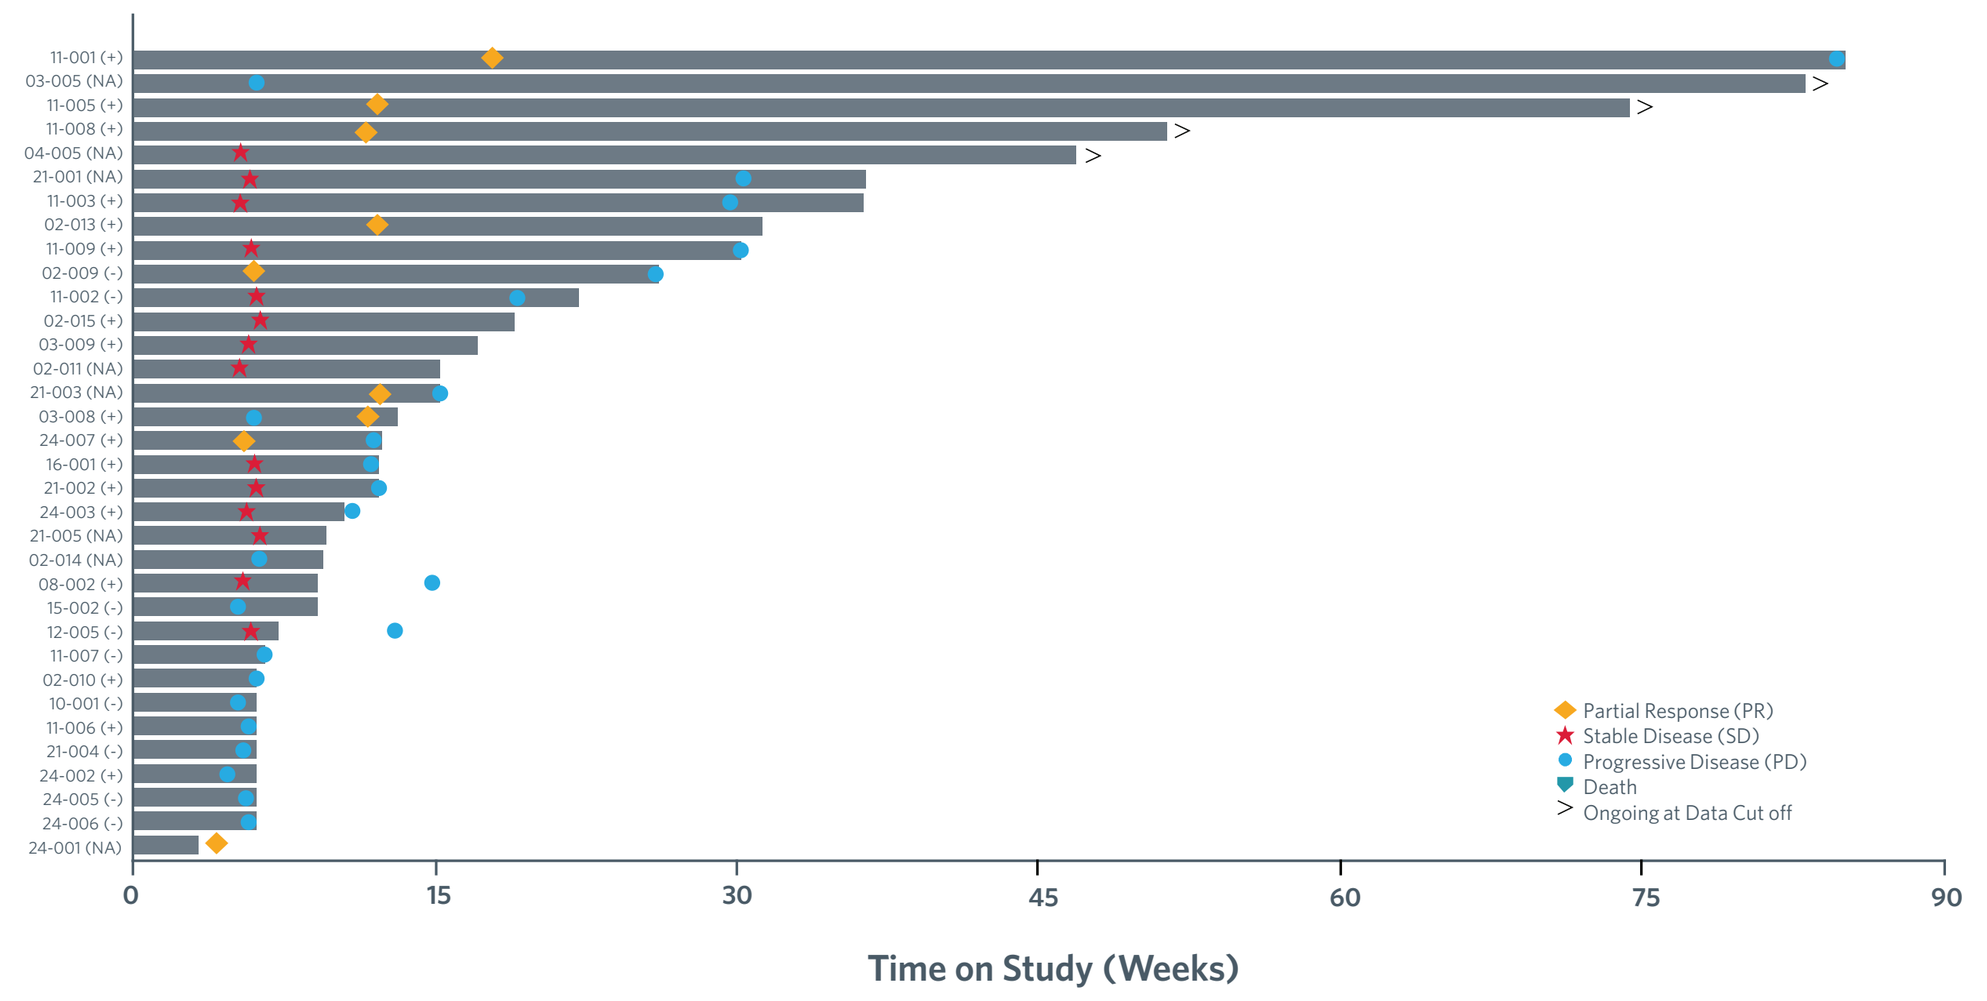
Partial Response (PR)
Stable Disease (SD)
Progressive Disease (PD)
Death
Ongoing at Data Cut off
0
15
30
45
60
75
90
Time on Study (Weeks)

(+/-) next to patient number denotes PD-L1 expression.

Figure 4. **Change in Tumor Size from Baseline Over Time.** Durable responses were observed.

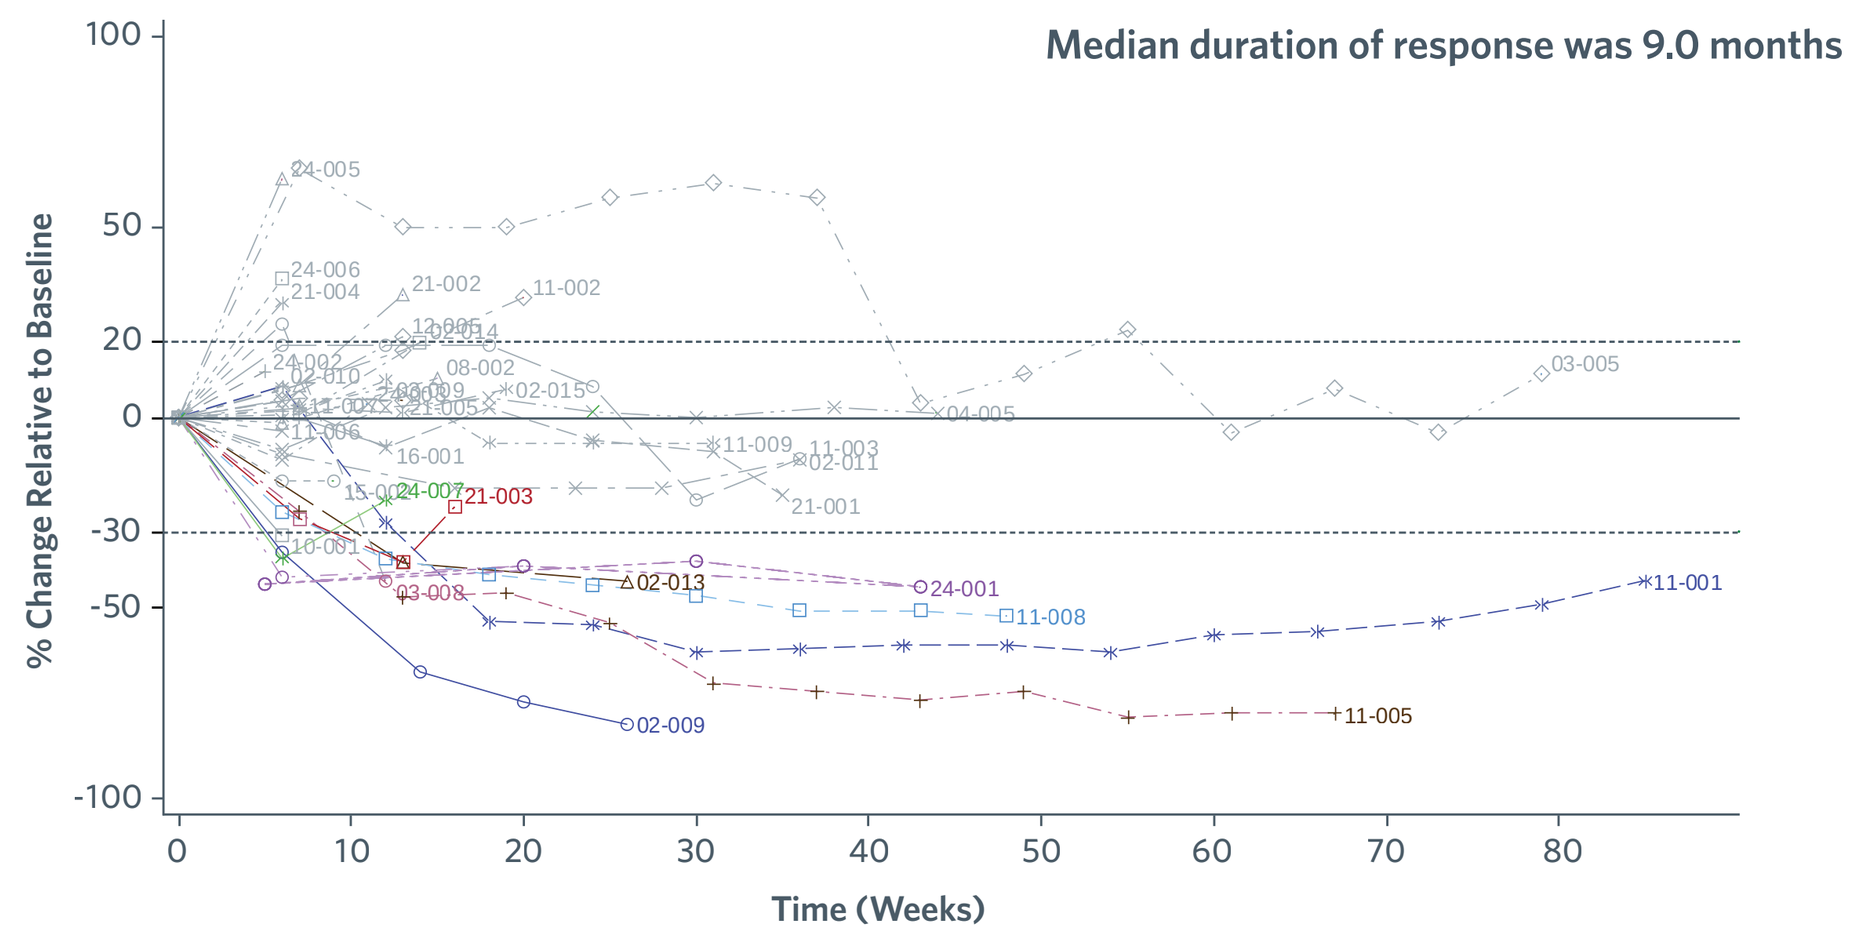
Median duration of response was 9.0 months
% Change Relative to Baseline
100
50
20
0
-30
-50
-100
0
10
20
30
40
50
60
70
80
Time (Weeks)
02-009
11-005
11-001
11-008
24-001
02-013
03-005

EFFICACY (continued)

Table 3. **Characteristics of Patients with Confirmed Partial Response.** Six confirmed partial responses have been observed, the majority were PD-L1 positive by fresh biopsy. Two of the six patients are ongoing in the study.

Patient	Baseline PD-L1 Expression	Prior Anti-PD-1	Best Response to Prior Anti-PD-1 (mo)	Duration on Prior Tx (mo)	Time Since Last Dose Prior Anti-PD-1 (mo)	Duration of Response on Trial (mo)	Status at Time of Data Cutoff
11-001	+	Nivolumab	SD	12.5	28.8	5.6	Discontinued for PD
11-005	+	Nivolumab	PD	3.3	1.8	14.5+	Ongoing
11-008	+	Pembrolizumab	SD	23.1	6.2	9.3+	Ongoing
02-013	+	Pembrolizumab	SD	19.0	1.2	4.5	Discontinued
02-009	-	Nivolumab	SD	6.5	10.4	4.7	Discontinued for PD
24-001	NA	Pembrolizumab	PD	2.8	4.3	8.9	Discontinued

CORRELATES

NanoString and Tumor Inflammation Signature Score

- Pre-treatment and post-treatment biomarker analysis included gene expression by a Beta version of the PanCancer IO 360™ assay (NanoString® Technologies, Inc, Seattle, WA).
- This assay is for research use only and is not for use in diagnostic procedures. NanoString is a registered trademark of NanoString Technologies, Inc., in the United States and/or other countries.
- Tumor Inflammation Signature (TIS) score was calculated as a linear combination of 18 genes that measure peripheral suppressed immune response and are normalized to 10 housekeeping genes previously identified to have stable expression across all cancer types.[6]

Figure 5. **Gene Expression was Assessed Pre- and Post-treatment.** Several pathways associated with inflammation increased with treatment. Increased immune function signatures were observed consistently in samples from responders.

A) Change in Signature Analysis from Screen to C2D15

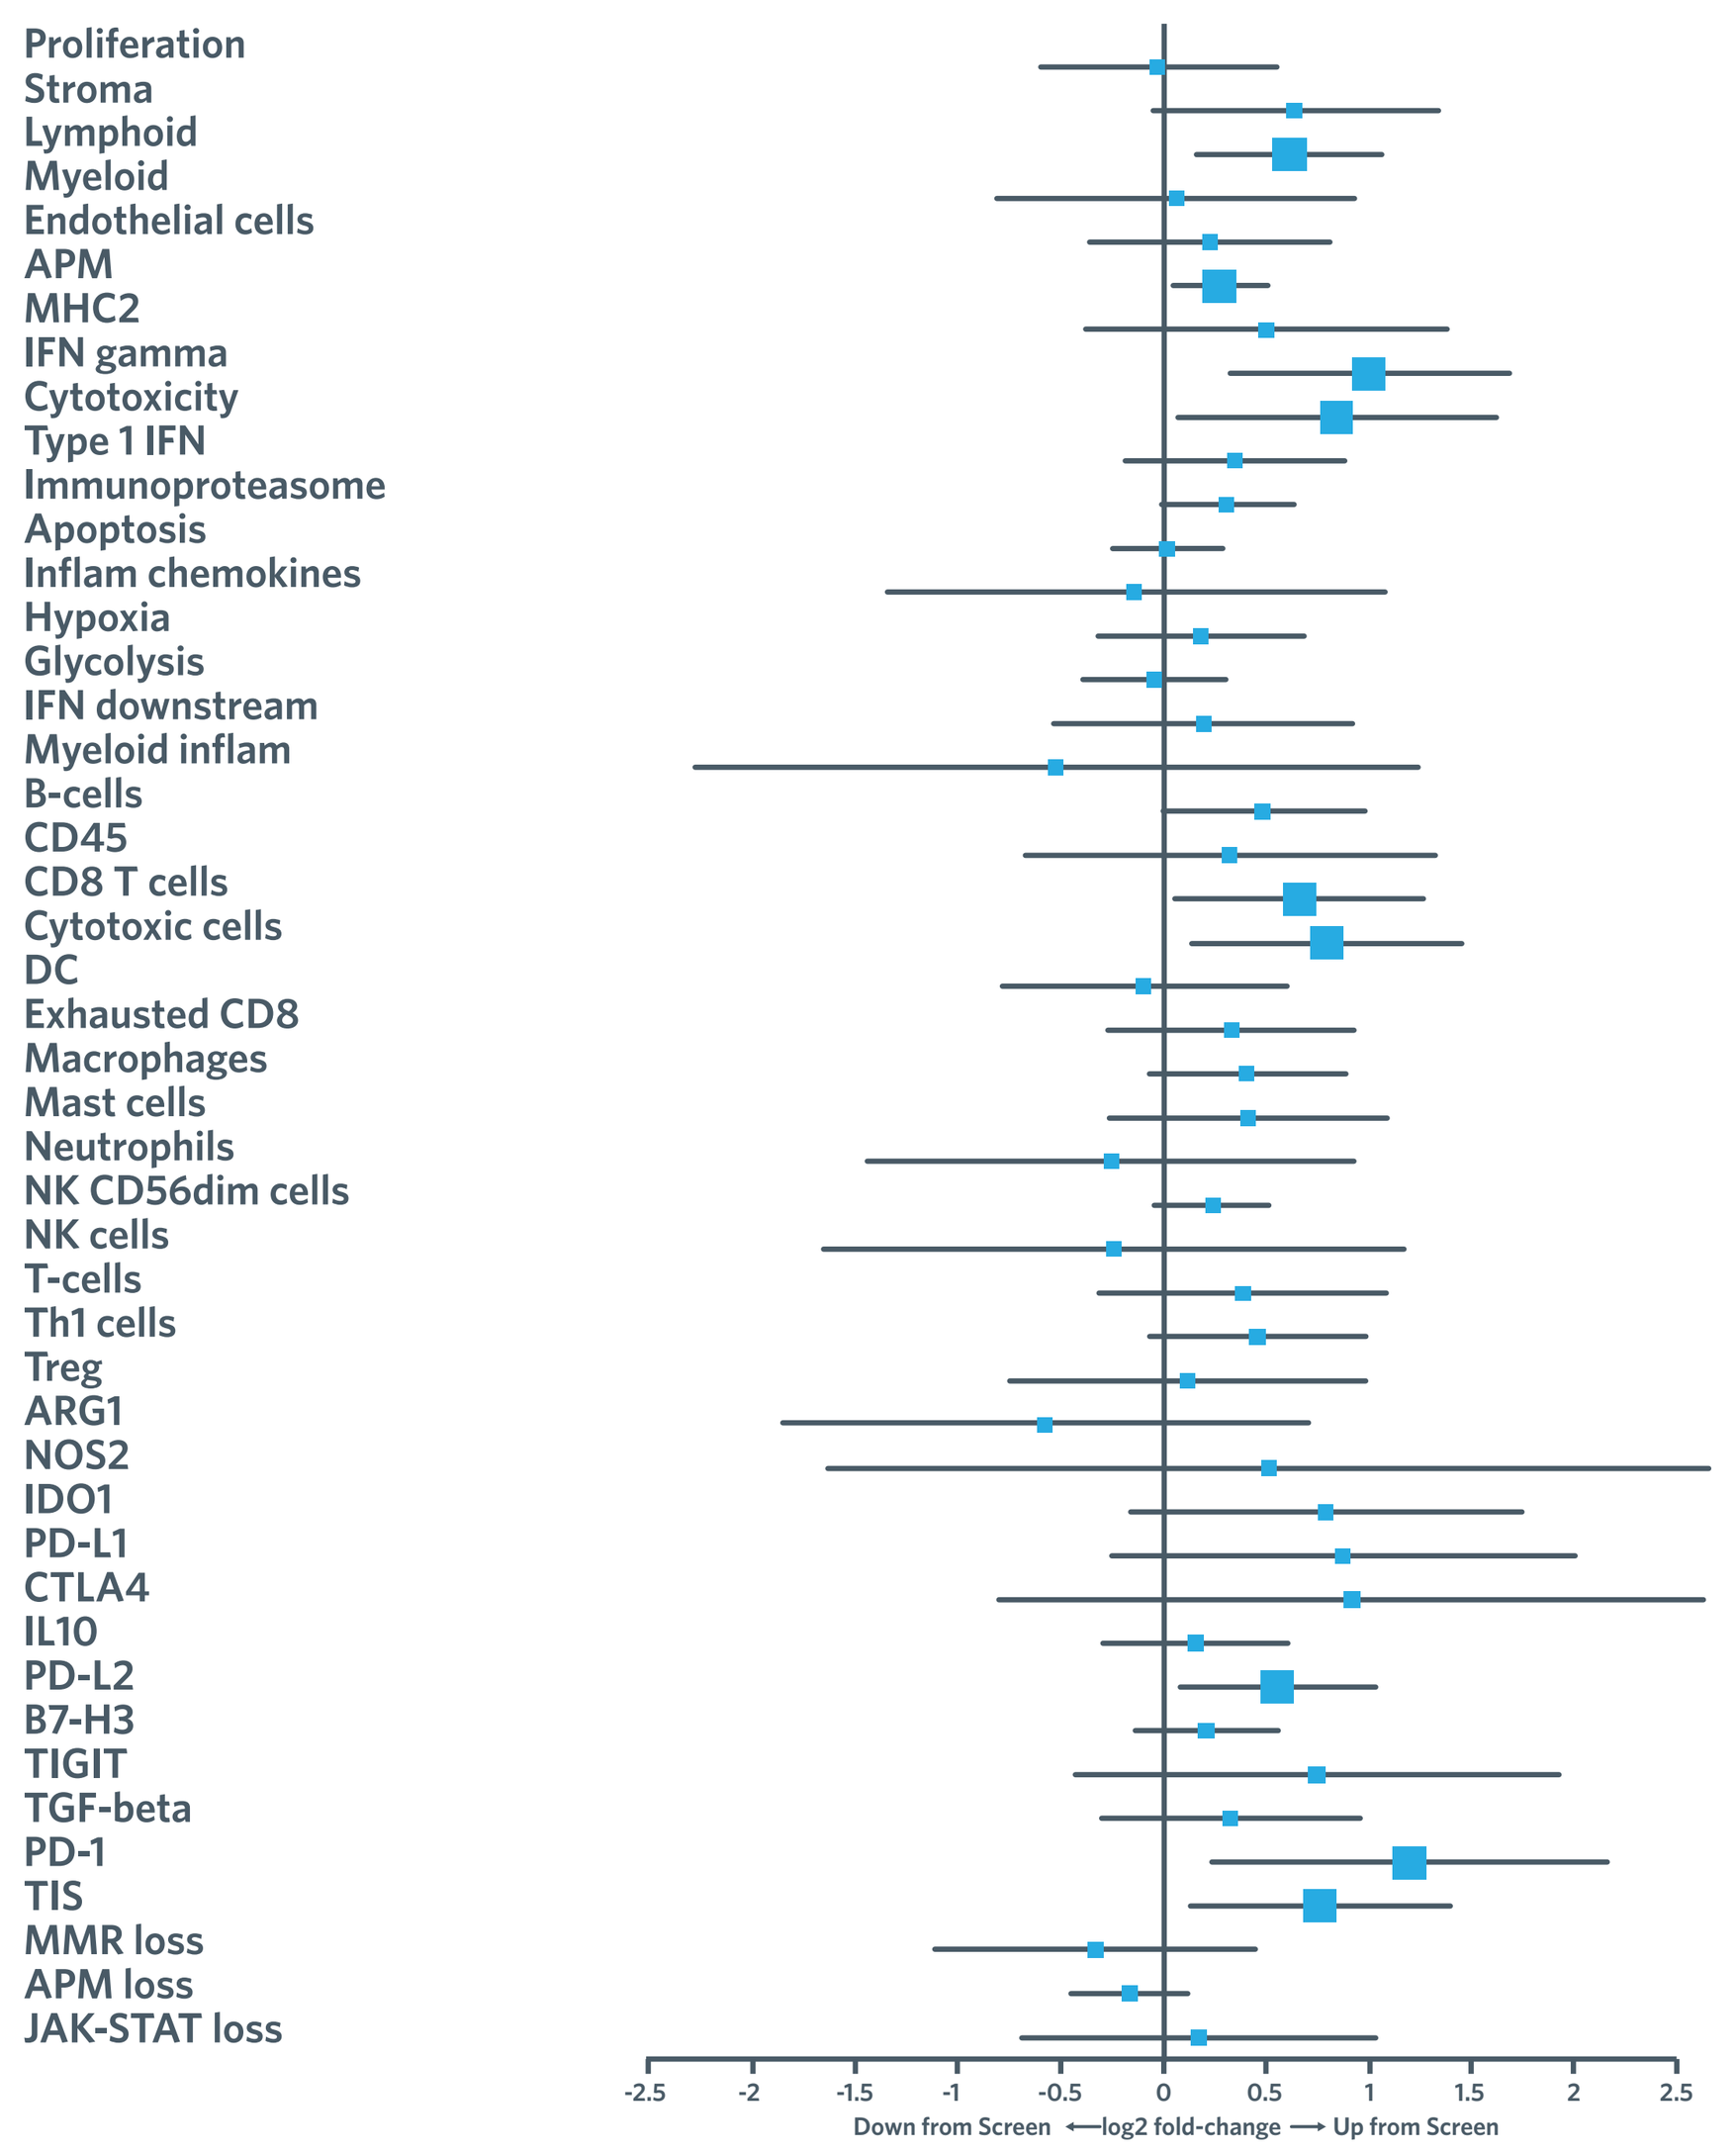
Proliferation
Stroma
Lymphoid
Myeloid
Endothelial cells
APM
MHC2
IFN gamma
Cytotoxicity
Type 1 IFN
Immunoproteasome
Apoptosis
Inflam chemokines
Hypoxia
Glycolysis
IFN downstream
Myeloid inflam
B-cells
CD45
CD8 T cells
Cytotoxic cells
DC
Exhausted CD8
Macrophages
Mast cells
Neutrophils
NK CD56dim cells
NK cells
T-cells
Th1 cells
Treg
ARG1
NOS2
IDO1
PD-L1
CTLA4
IL10
PD-L2
B7-H3
TIGIT
TGF-beta
PD-1
TIS
MMR loss
APM loss
JAK-STAT loss
-2.5
-2
-1.5
-1
-0.5
0
0.5
1
1.5
2
2.5
Down from Screen ⟵log2 fold-change⟶ Up from Screen

CORRELATES (continued)

B) Immune Function Plot

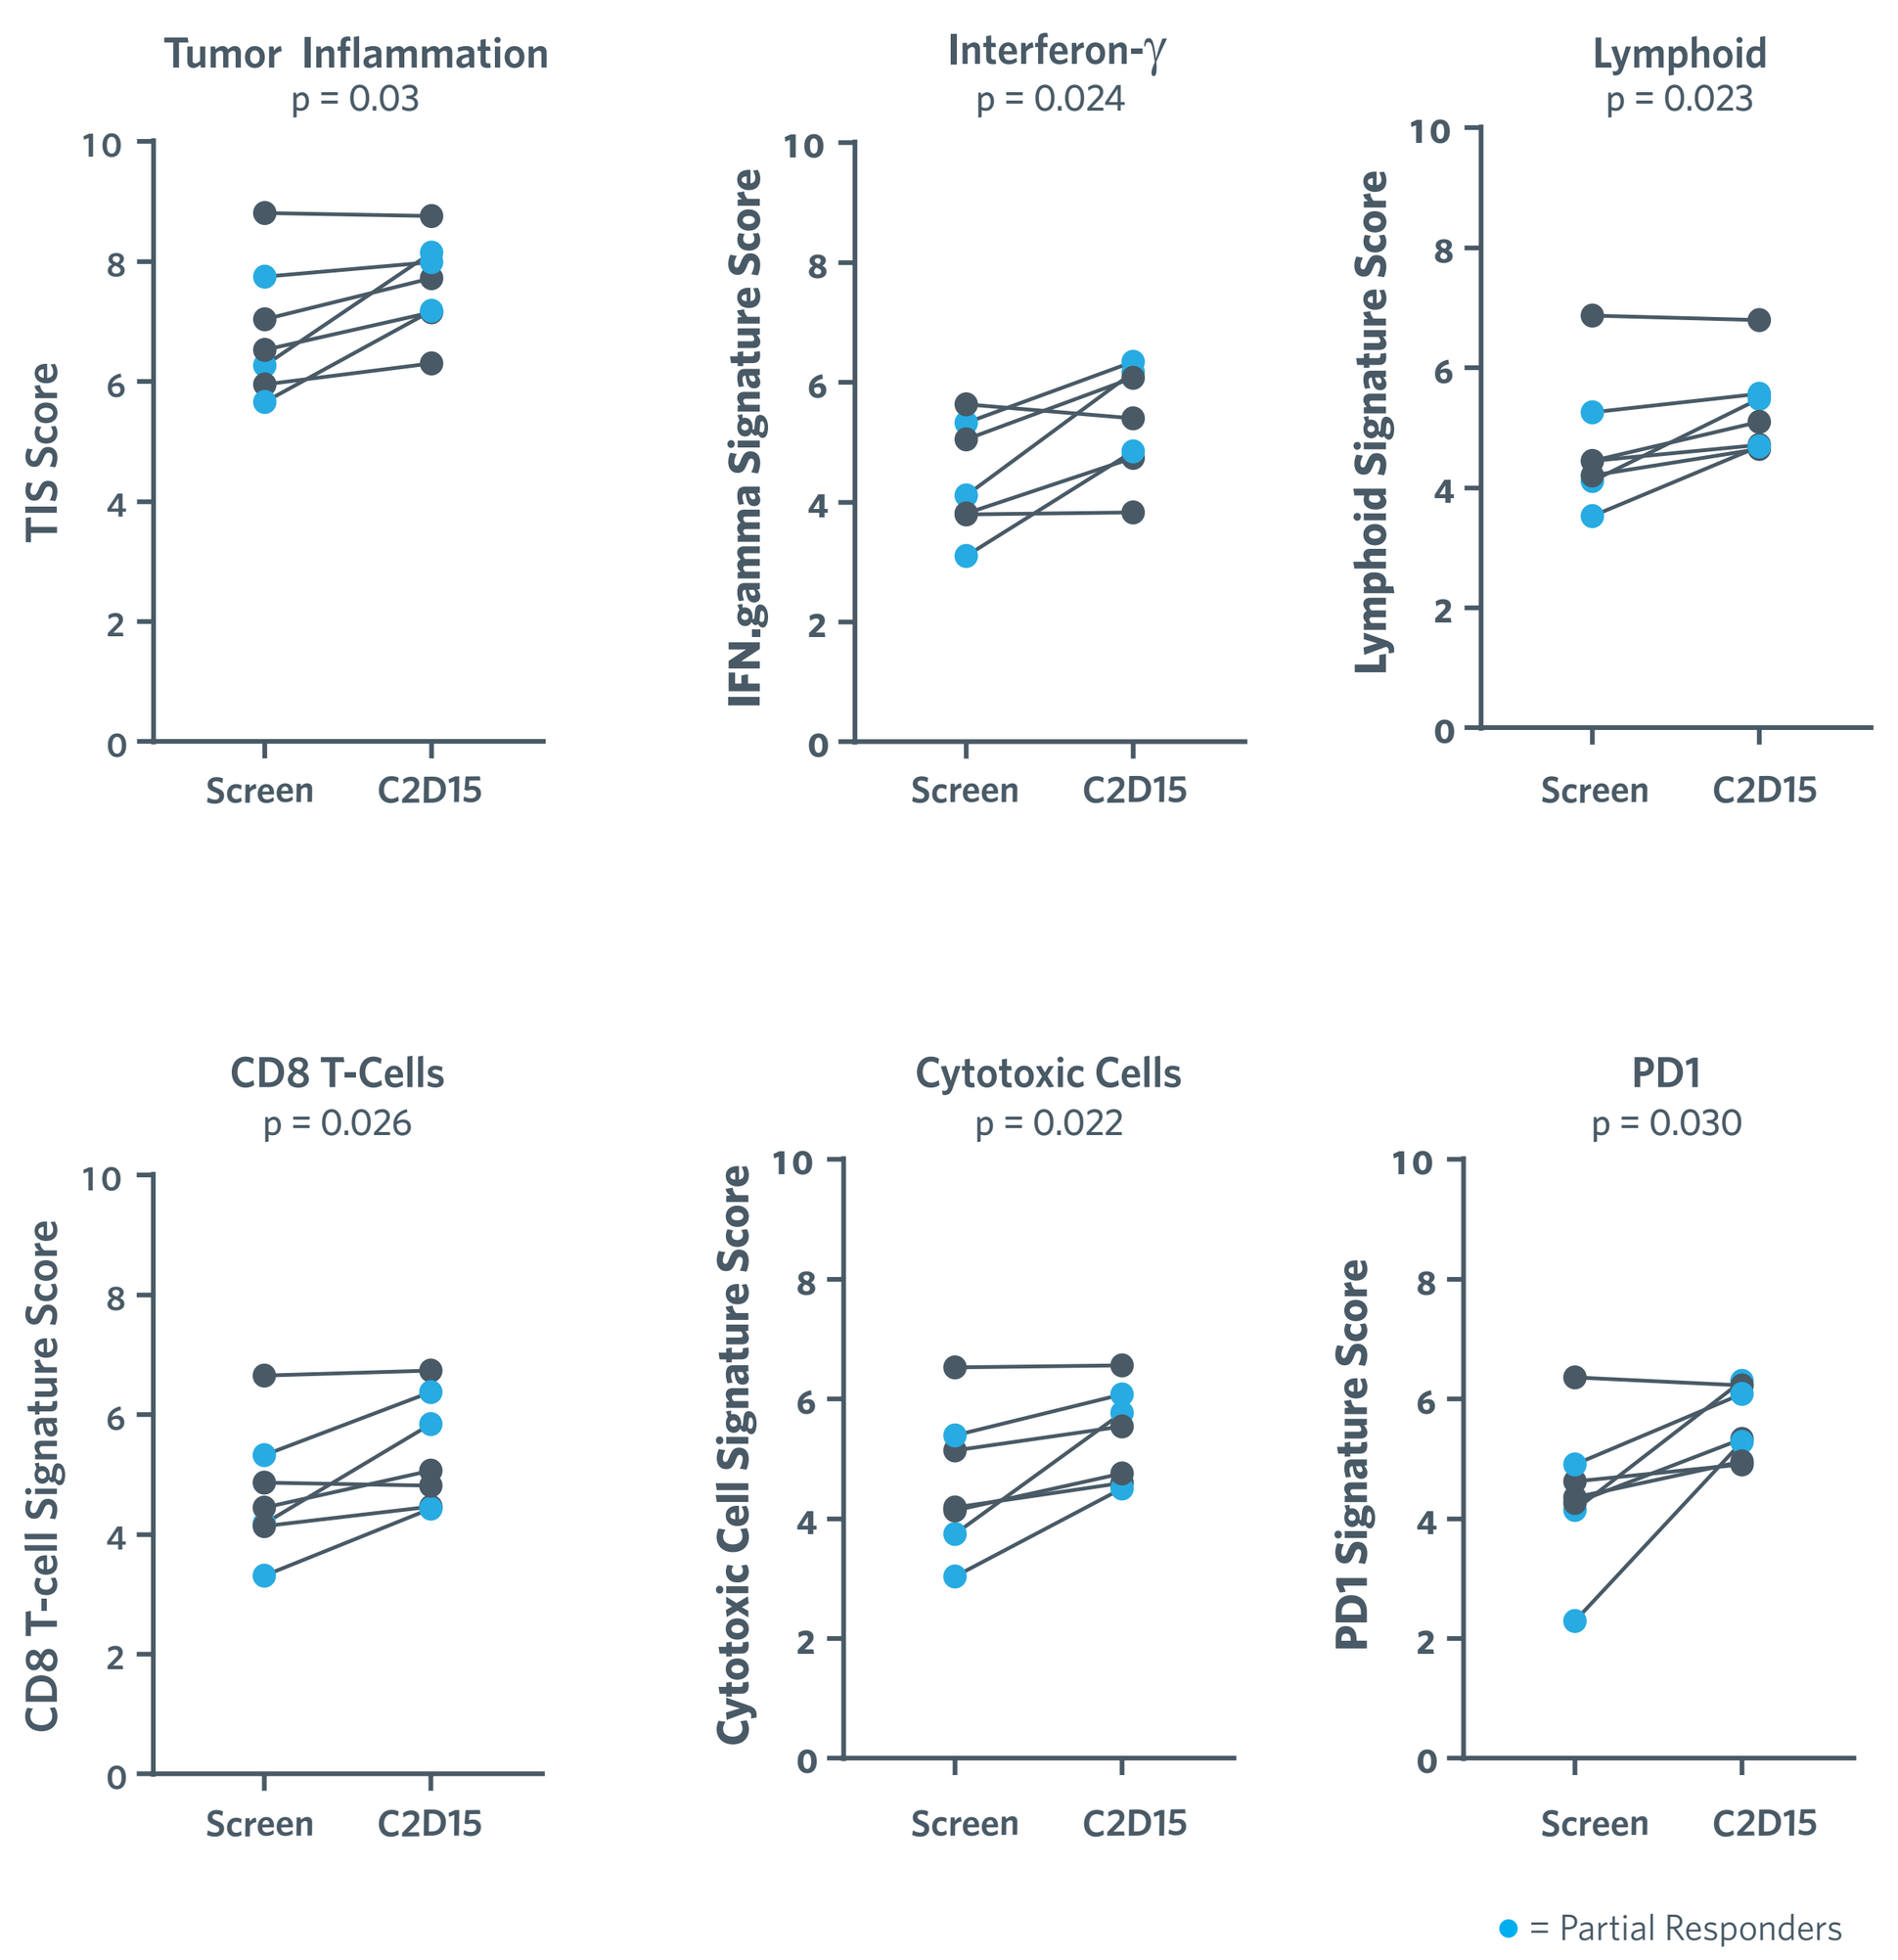
Tumor Inflammation
p = 0.03
TIS Score
Interferon-γ
p = 0.024
IFN.gamma Signature Score
Lymphoid
p = 0.023
Lymphoid Signature Score
CD8 T-Cells
p = 0.026
CD8 T-cell Signature Score
Cytotoxic Cells
p = 0.022
Cytotoxic Cell Signature Score
PD1
p = 0.030
PD1 Signature Score
Screen
C2D15
= Partial Responders

- Tumor biopsy samples were captured at screening and C2D15 of treatment and gene expression was assessed on the NanoString PanCancer IO 360™ panel.
- Signature analysis reveals that several pathways associated with inflammation are upregulated **(Figure 5A)** after treatment.
- Signatures specific to increased immune function were among the strongest signals that were observed **(Figure 5B)**. This was consistently observed in samples from patients with partial responses **(Blue Symbols)**.
- Together, these data suggest that treatment with entinostat/pembrolizumab increases inflammation and supports immune function within the tumor microenvironment.
- Based on results seen with entinostat and pembrolizumab in NSCLC[7], analyses are ongoing to assess the potential utility of baseline classical monocytes as a biomarker in melanoma and other tumor types.

SAFETY

- Table 4 shows treatment-emergent adverse events (TEAEs) related to entinostat or pembrolizumab or both observed in ≥10% of patients at any grade, or in any patients at grade ≥3.
- TEAEs grade 3 or higher related to study treatment occurred in 14/34 patients.
 - Two patients experienced Grade 3/4 irAEs (1 event each of colitis and pneumonitis). An additional 12 patients experienced other Grade 3/4 AEs with only anemia, rash, neutropenia, and hyponatremia occurring in two or more patients.
- 4 (12%) patients discontinued due to a treatment-related AE. These events included liver enzyme increase, mucosal inflammation, colitis, and autoimmune hepatitis.

SAFETY (continued)

Table 4: Safety	Any Grade (N=34)	Grade ≥3 (N=34)
Subjects With At Least One Treatment-Emergent Adverse Event Related to Study Treatment	**30 (88.2)**	**14 (41.2)**
Gastrointestinal disorders		
Nausea	18 (52.9)	
Diarrhea	7 (20.6)	
Colitis	1 (2.9)	
Stomatitis	1 (2.9)	
General disorders and administration site conditions		
Fatigue	12 (35.3)	1 (2.9)
Mucosal inflammation	1 (2.9)	1 (2.9)
Respiratory, thoracic and mediastinal disorders		
Dyspnea	3 (8.8)	
Pneumonitis	1 (2.9)	1 (2.9)
Skin and subcutaneous tissue disorders		
Pruritus	6 (17.6)	
Rash	4 (11.8)	2 (5.9)
Investigations		
Neutrophil count decreased	5 (14.7)	
Platelet count decreased	4 (11.8)	
Alanine aminotransferase increased	2 (5.9)	1 (2.9)
Aspartate aminotransferase increased	2 (5.9)	1 (2.9)
Blood alkaline phosphatase increased	1 (2.9)	1 (2.9)
Blood bilirubin increased	1 (2.9)	1 (2.9)
Metabolism and nutrition disorders		
Dehydration	2 (5.9)	
Hyponatremia	2 (5.9)	2 (5.9)
Hyperglycemia	1 (2.9)	1 (2.9)
Musculoskeletal and connective tissue disorders		
Myalgia	4 (11.8)	
Back pain	2 (5.9)	
Pain in extremity	1 (2.9)	1 (2.9)
Cardiac disorders		
Atrial fibrilation	1 (2.9)	1 (2.9)
Blood and lymphatic system disorders		
Anemia	4 (11.8)	2 (5.9)
Neutropenia	3 (8.8)	2 (5.9)

CONCLUSIONS

- ENT + PEMBRO continues to demonstrate promising anti-tumor activity and acceptable safety in patients with melanoma who have progressed on prior PD-(L)1 blockade.
- Preliminary biomarker analysis supports the hypothesis that the addition of ENT increases inflammation in the tumor microenvironment necessary for successful re-treatment with an anti–PD-(L)1.
- Additional analyses are ongoing to identify biomarkers that can predict clinical benefit in this population.
- Data in the study continues to mature, with enrollment now complete (n=55).

References:

1. Zimmer L, et al. *Eur J Cancer.* 2017;75:47-55.
2. Orillion A, et al. *Clin Cancer Res.* 2017;23:5187-5201.
3. Johnson ML, et al. Presented at the ASCO Annual Meeting, 2017. Abstract 9529.
4. Johnson ML, et al. Presented at SITC Annual Meeting, 2017. Abstract P72.
5. Gandhi L, et al. Presented at SITC Annual Meeting, 2017. Abstract O19.
6. Ayers et al. *J Clin Invest*. 2017;127:2930-2940.
7. Gandhi L, et al. Presented at the ASCO Annual Meeting, 2018. Abstract 9036.

Acknowledgments

The authors thank the patients and their families, investigators and study staff. We acknowledge NanoString® Technologies, Inc. for providing the PanCancer IO 360™ panels for this work. We also thank Amy Sullivan, Sarah Church, Elena Viboch of the Nanostring Team for their contributions to this analysis and our ongoing collaboration. This study was sponsored by Syndax Pharmaceuticals, Inc., in collaboration with Merck & Co., Inc., Kenilworth, NJ. Writing and editorial assistance was provided by BluPrint Oncology, LLC. This assistance was funded by Syndax Pharmaceuticals, Inc.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® and the author of this poster.

Poster presented at the American Society of Clinical Oncology Annual Meeting, June 1-5, 2018, Chicago IL.