P72

Analysis of Biomarkers From a Cohort of Advanced Melanoma Patients Treated With Entinostat (ENT) and Pembrolizumab (PEMBRO) Previously Exposed to Immune Checkpoint Inhibition

Melissa L. Johnson,[1] Dmitry Gabrilovich,[2] Fang Wang,[2] Rene Gonzalez,[3] Mateusz Opyrchal,[4] David Tamang,[5] Emmett Schmidt,[6] Michael L. Meyers,[5] Peter Ordentlich,[5] Sanjiv S. Agarwala[7]

[1]Sarah Cannon Research Institute, Nashville, TN; [2]The Wistar Institute, Philadelphia, PA; [3]University of Colorado Cancer Center, Aurora, CO; [4]Roswell Park Cancer Institute, Buffalo, NY; [5]Syndax Pharmaceuticals, Inc, Waltham, MA; [6]Merck & Co, Inc, Kenilworth, NJ; [7]St. Luke's Cancer Center and University Health Network, Easton, PA

BACKGROUND

- Although immune checkpoint inhibition targeting cytotoxic T lymphocyte antigen 4 (CTLA-4) and/or programmed cell death-1 (PD-1) has significantly improved survival outcomes for advanced melanoma patients, a large segment of the patient population does not respond or experiences recurrence after a relatively short duration of treatment[1]
- Resistance to mounting an effective antitumor immune response is multifactorial (**Figure 1**), and a multitude of combination approaches are being tested to address the problem[1]

Figure 1. **Differences Between Tumors With "Inflamed" and "Non-Inflamed" Immunophenotypes and Potential Therapeutic Interventions[2]**

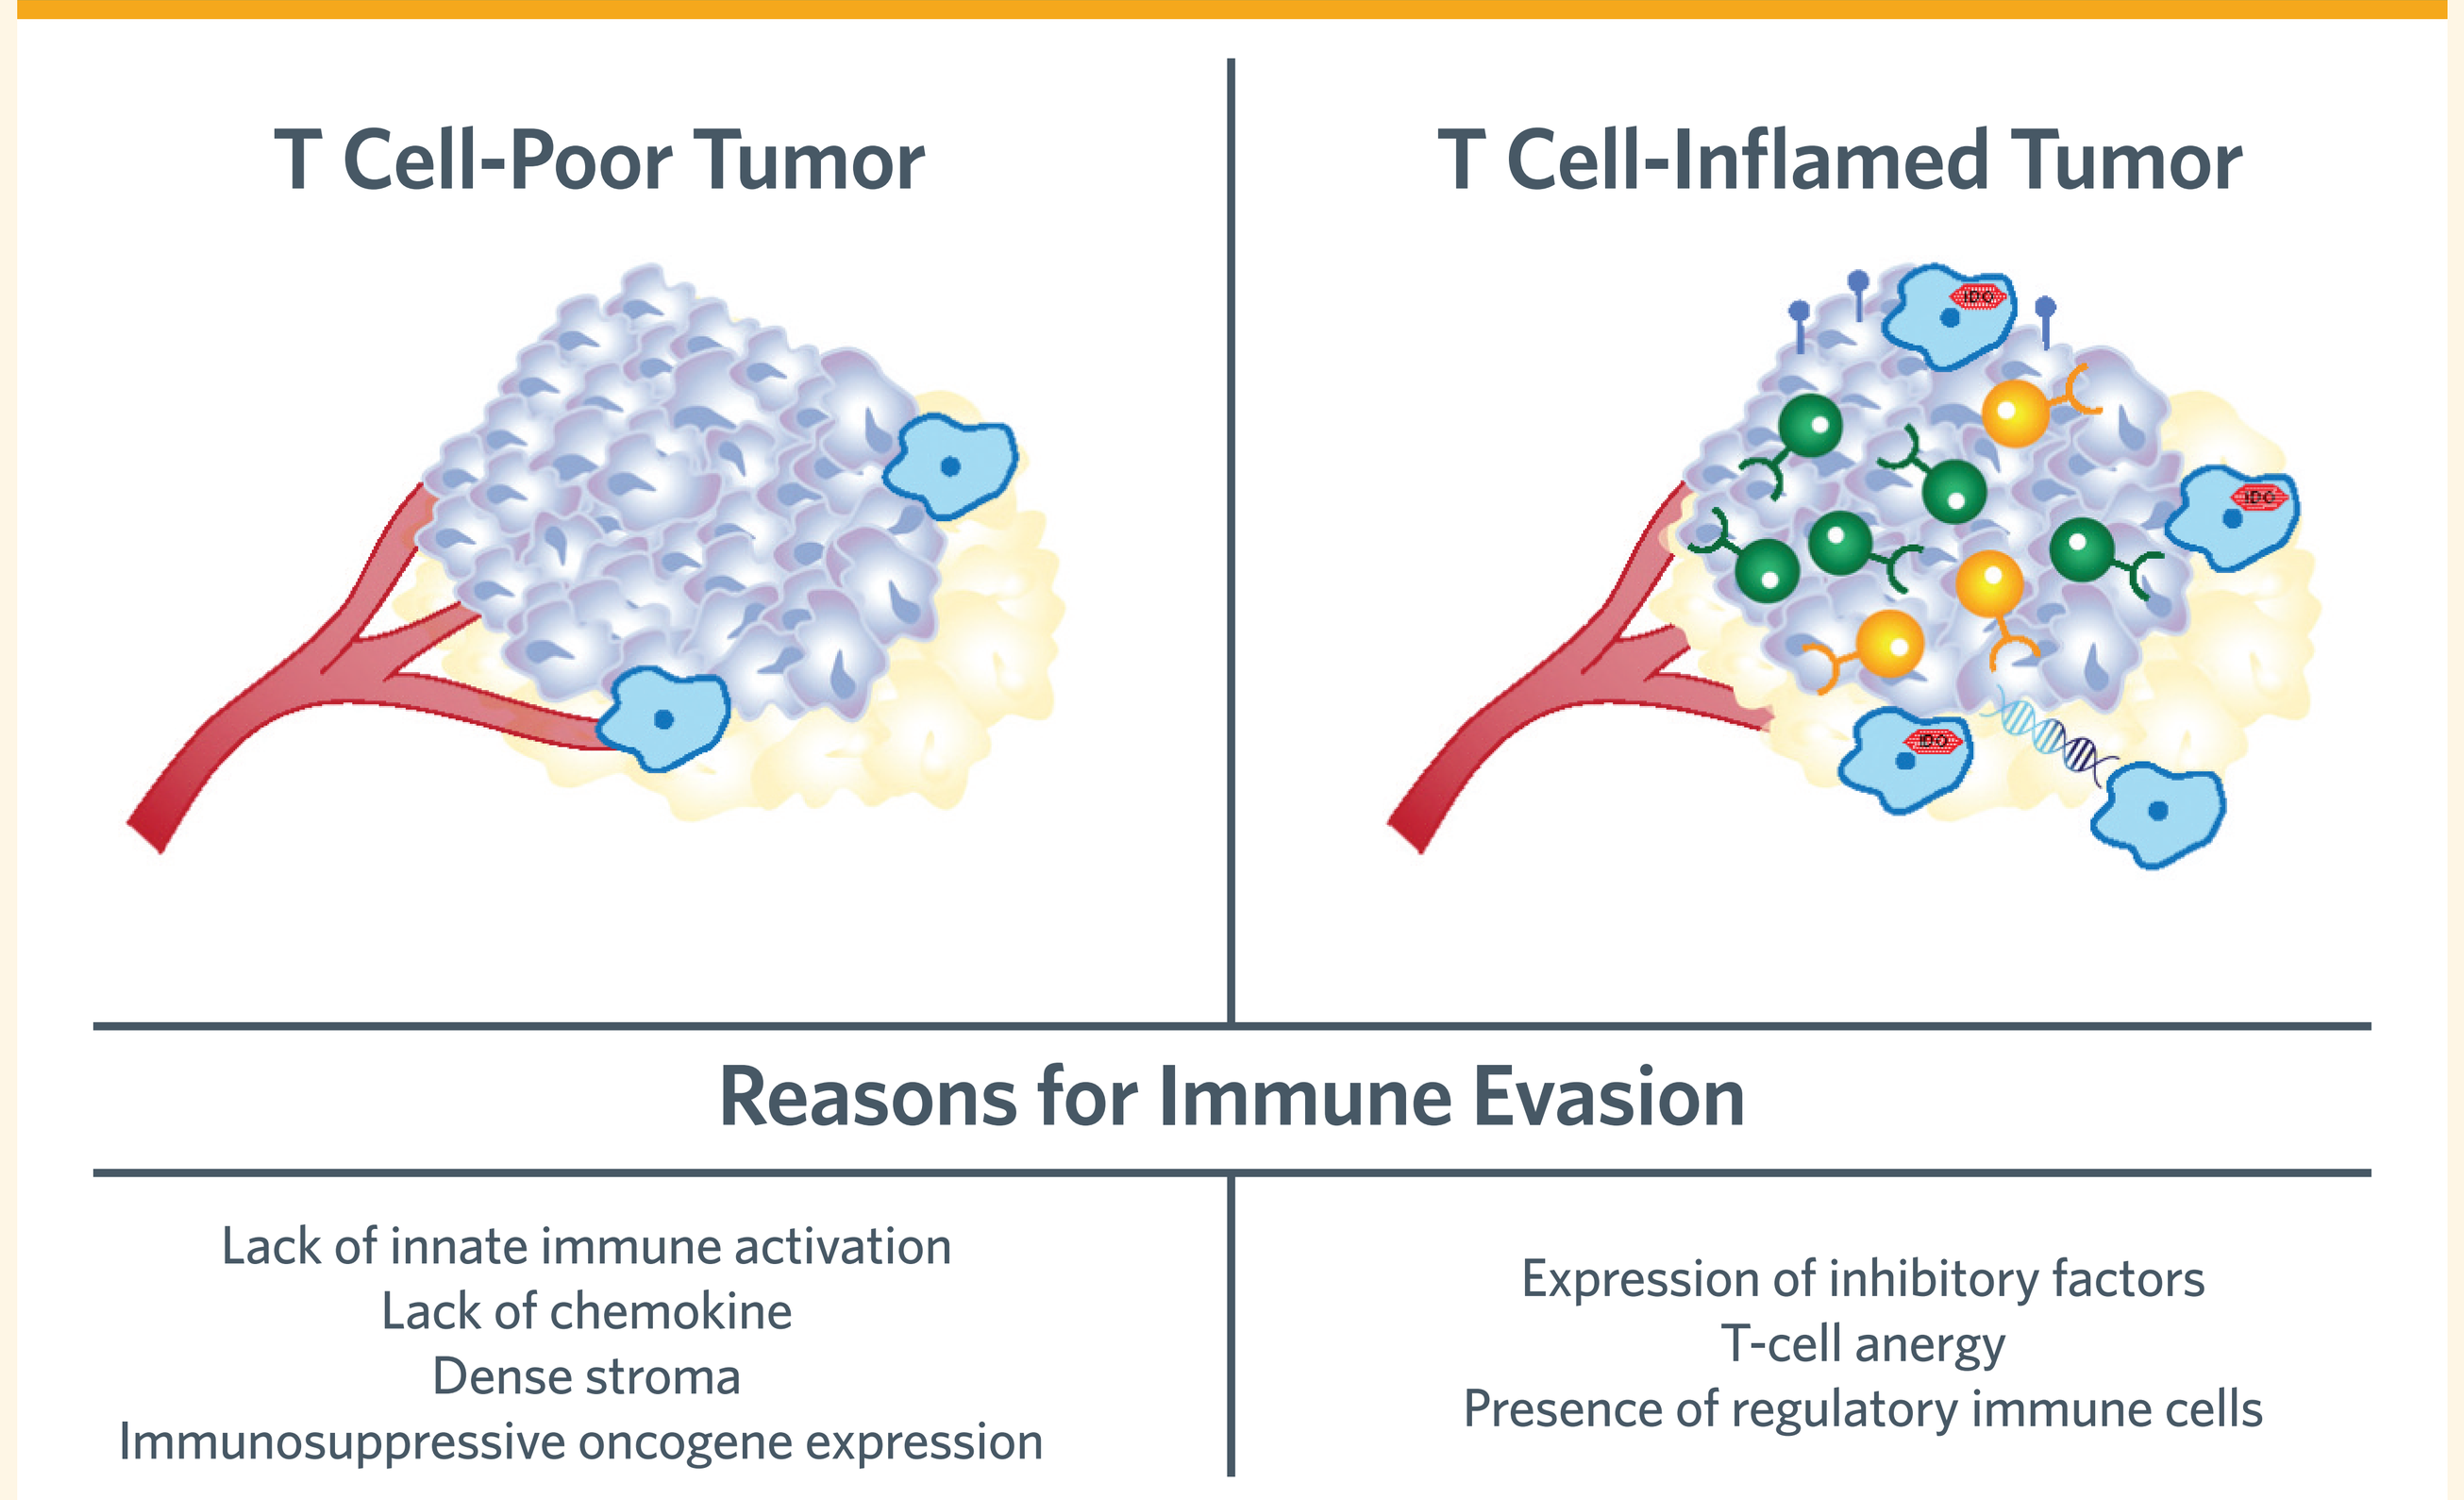
T Cell-Poor Tumor
T Cell-Inflamed Tumor
Reasons for Immune Evasion
Lack of innate immune activation
Lack of chemokine
Dense stroma
Immunosuppressive oncogene expression
Expression of inhibitory factors
T-cell anergy
Presence of regulatory immune cells

Adapted under a Creative Commons license from Spranger S, Gajewski T. *J Immunother Cancer.* 2013;1:16.

- Entinostat (ENT) is an oral, class 1 selective histone deacetylase inhibitor that has been shown in a preclinical study[1] to synergize with immune checkpoint blockade to inhibit tumor growth through down-regulation of regulatory T cells (Treg) and myeloid-derived suppressor cells (MDSCs), functional immunosuppression, and alteration of an immune suppressive tumor microenvironment to one more favorable for antitumor immune response[1]
- Results from the ongoing ENCORE-601 study demonstrated that ENT combined with pembrolizumab (PEMBRO) elicited a 31% response rate in a cohort of 13 patients with melanoma progressing on or after a PD-1 blocking antibody[3]
- Correlative study analyses of pre- and posttreatment tissue and blood samples from patients treated with ENT plus PEMBRO have been conducted to confirm the mechanism of action of ENT in combination with immune checkpoint blockade and whether predictive markers of response can be identified[3]

METHODS

- ENCORE-601 employed a Simon 2-stage design[4] to assess activity of ENT 5 mg QW PO, combined with PEMBRO 200 mg Q3W IV across 4 cohorts:
 1. advanced anti-PD-(ligand [L])1–naive non-small cell lung cancer (NSCLC);
 2. advanced NSCLC progressed on anti–PD-(L)1 treatment;
 3. advanced melanoma progressed on anti–PD-(L)1 treatment;
 4. advanced anti-PD-(L)1–naive microsatellite stable colorectal cancer

Key Exploratory Objectives

- To assess the ratio of effector T cells to Tregs in peripheral blood and tumor biopsies pre- and posttherapy
- To evaluate inflammatory T-cell signature changes in peripheral blood and tumor biopsies pre- and posttherapy
- To evaluate changes in number of MDSCs in peripheral blood and tumor biopsies pre- and posttherapy (flow cytometry)
- Pretreatment and posttreatment biomarker analysis included gene expression by a beta version of the PanCancer IO 360™ assay (NanoString® Technologies, Inc, Seattle, WA), PD-(L)1 expression by immunohistochemistry and levels of CD8+ T cells and LOX1+CD15+MDSCs by immunofluorescence staining on paraffin-embedded human tissue sections. Phenotypic evaluation of immune cell subsets was conducted in peripheral blood samples collected pretreatment (cycle 2, day 1 [C2D1]) and posttreatment (C2D15)
 — FOR RESEARCH USE ONLY. Not for use in diagnostic procedures. NanoString is a registered trademark of NanoString Technologies, Inc., in the United States and/or other countries
- Tumor Inflammation Signature (TIS) score was calculated as a linear combination of 18 genes that measure peripheral suppressed immune response and are normalized to 10 housekeeping genes previously identified to have stable expression across all cancer types[5]

RESULTS

Clinical Efficacy Summary

- Baseline demographic and prior therapy data are summarized in **Table 1A** and **Table 1B**

Table 1A. **Patient Baseline Demographics and Treatment History**

	Total (n=13)		Total (n=13)
Sex, n (%)		Baseline LDH (%> ULN), n (%)	
Male/Female	9 (69%)/4 (31%)	Yes/No	5 (38%)/8 (62%)
Age (years)		2 or more Prior PD-1/PD-L1 Therapy, n (%)	
Median (range)	62.0 (38–86)	Yes/No	2 (15%)/11 (85%)
Baseline ECOG Performance Score		Previously Treated with BRAF Inhibitor	
0/1	8 (62%)/5 (38%)	Yes/No	2 (15%)/11 (85%)
PD-L1 Expression, n (%)		Previously Treated with Ipilimumab & PD-1	
Negative	4 (31%)	Yes/No	8 (62%)/5 (38%)
Positive	6 (46%)	Previously Treated with Ipilimumab/Nivolumab	
Not Evaluable	3 (23%)	Yes/No	3 (23%)/10 (77%)

ECOG = Eastern Cooperative Oncology Group; LDH = lactate dehydrogenase; PD-(L)1 = programmed cell death-(ligand)1; UNL = upper limit of normal.

Table 1B. **Patient Prior History on PD-1 Therapy**

Patient ID	Best response to prior PD-1	Duration on to prior PD-1	Time between last PD-1 and 1st study dose	Best response on 601 study
02-011	PD	7.5 mo	0.7 mo	SD
15-002	PD	6.9 mo	2.1 mo	PD
11-006	PD	4.6 mo	2.1 mo	PD
11-005	PD	3.3 mo	1.8 mo	PR (Ongoing)
24-001	PD	2.8 mo	4.3 mo	PR
11-003	SD	20.3 mo	0.7 mo	SD (Ongoing)
11-001	SD	12.5 mo	28.8 mo	PR (Ongoing)
11-002	SD	12.0 mo	1.0 mo	SD
02-009	SD	6.5 mo	10.4 mo	PR
03-005	SD	4.9 mo	1.2 mo	SD (Ongoing)
10-001	SD	4.1 mo	1.3 mo	PD
16-001	SD	4.1 mo	1.3 mo	SD
02-010	CR	11.0 mo	12.4 mo	PD

CR = complete response; PD = progressive disease; PD-1 = programmed cell death-1; PR = partial response; SD = stable disease.

- 4 patients (31%) had a partial response (PR; 3 confirmed, 1 unconfirmed), and 4 (31%) additional patients had stable disease (**Figure 2**)
 — 4 patients continued on therapy with durable response/clinical benefit.

Figure 2. **Patient Response and Time on Treatment**

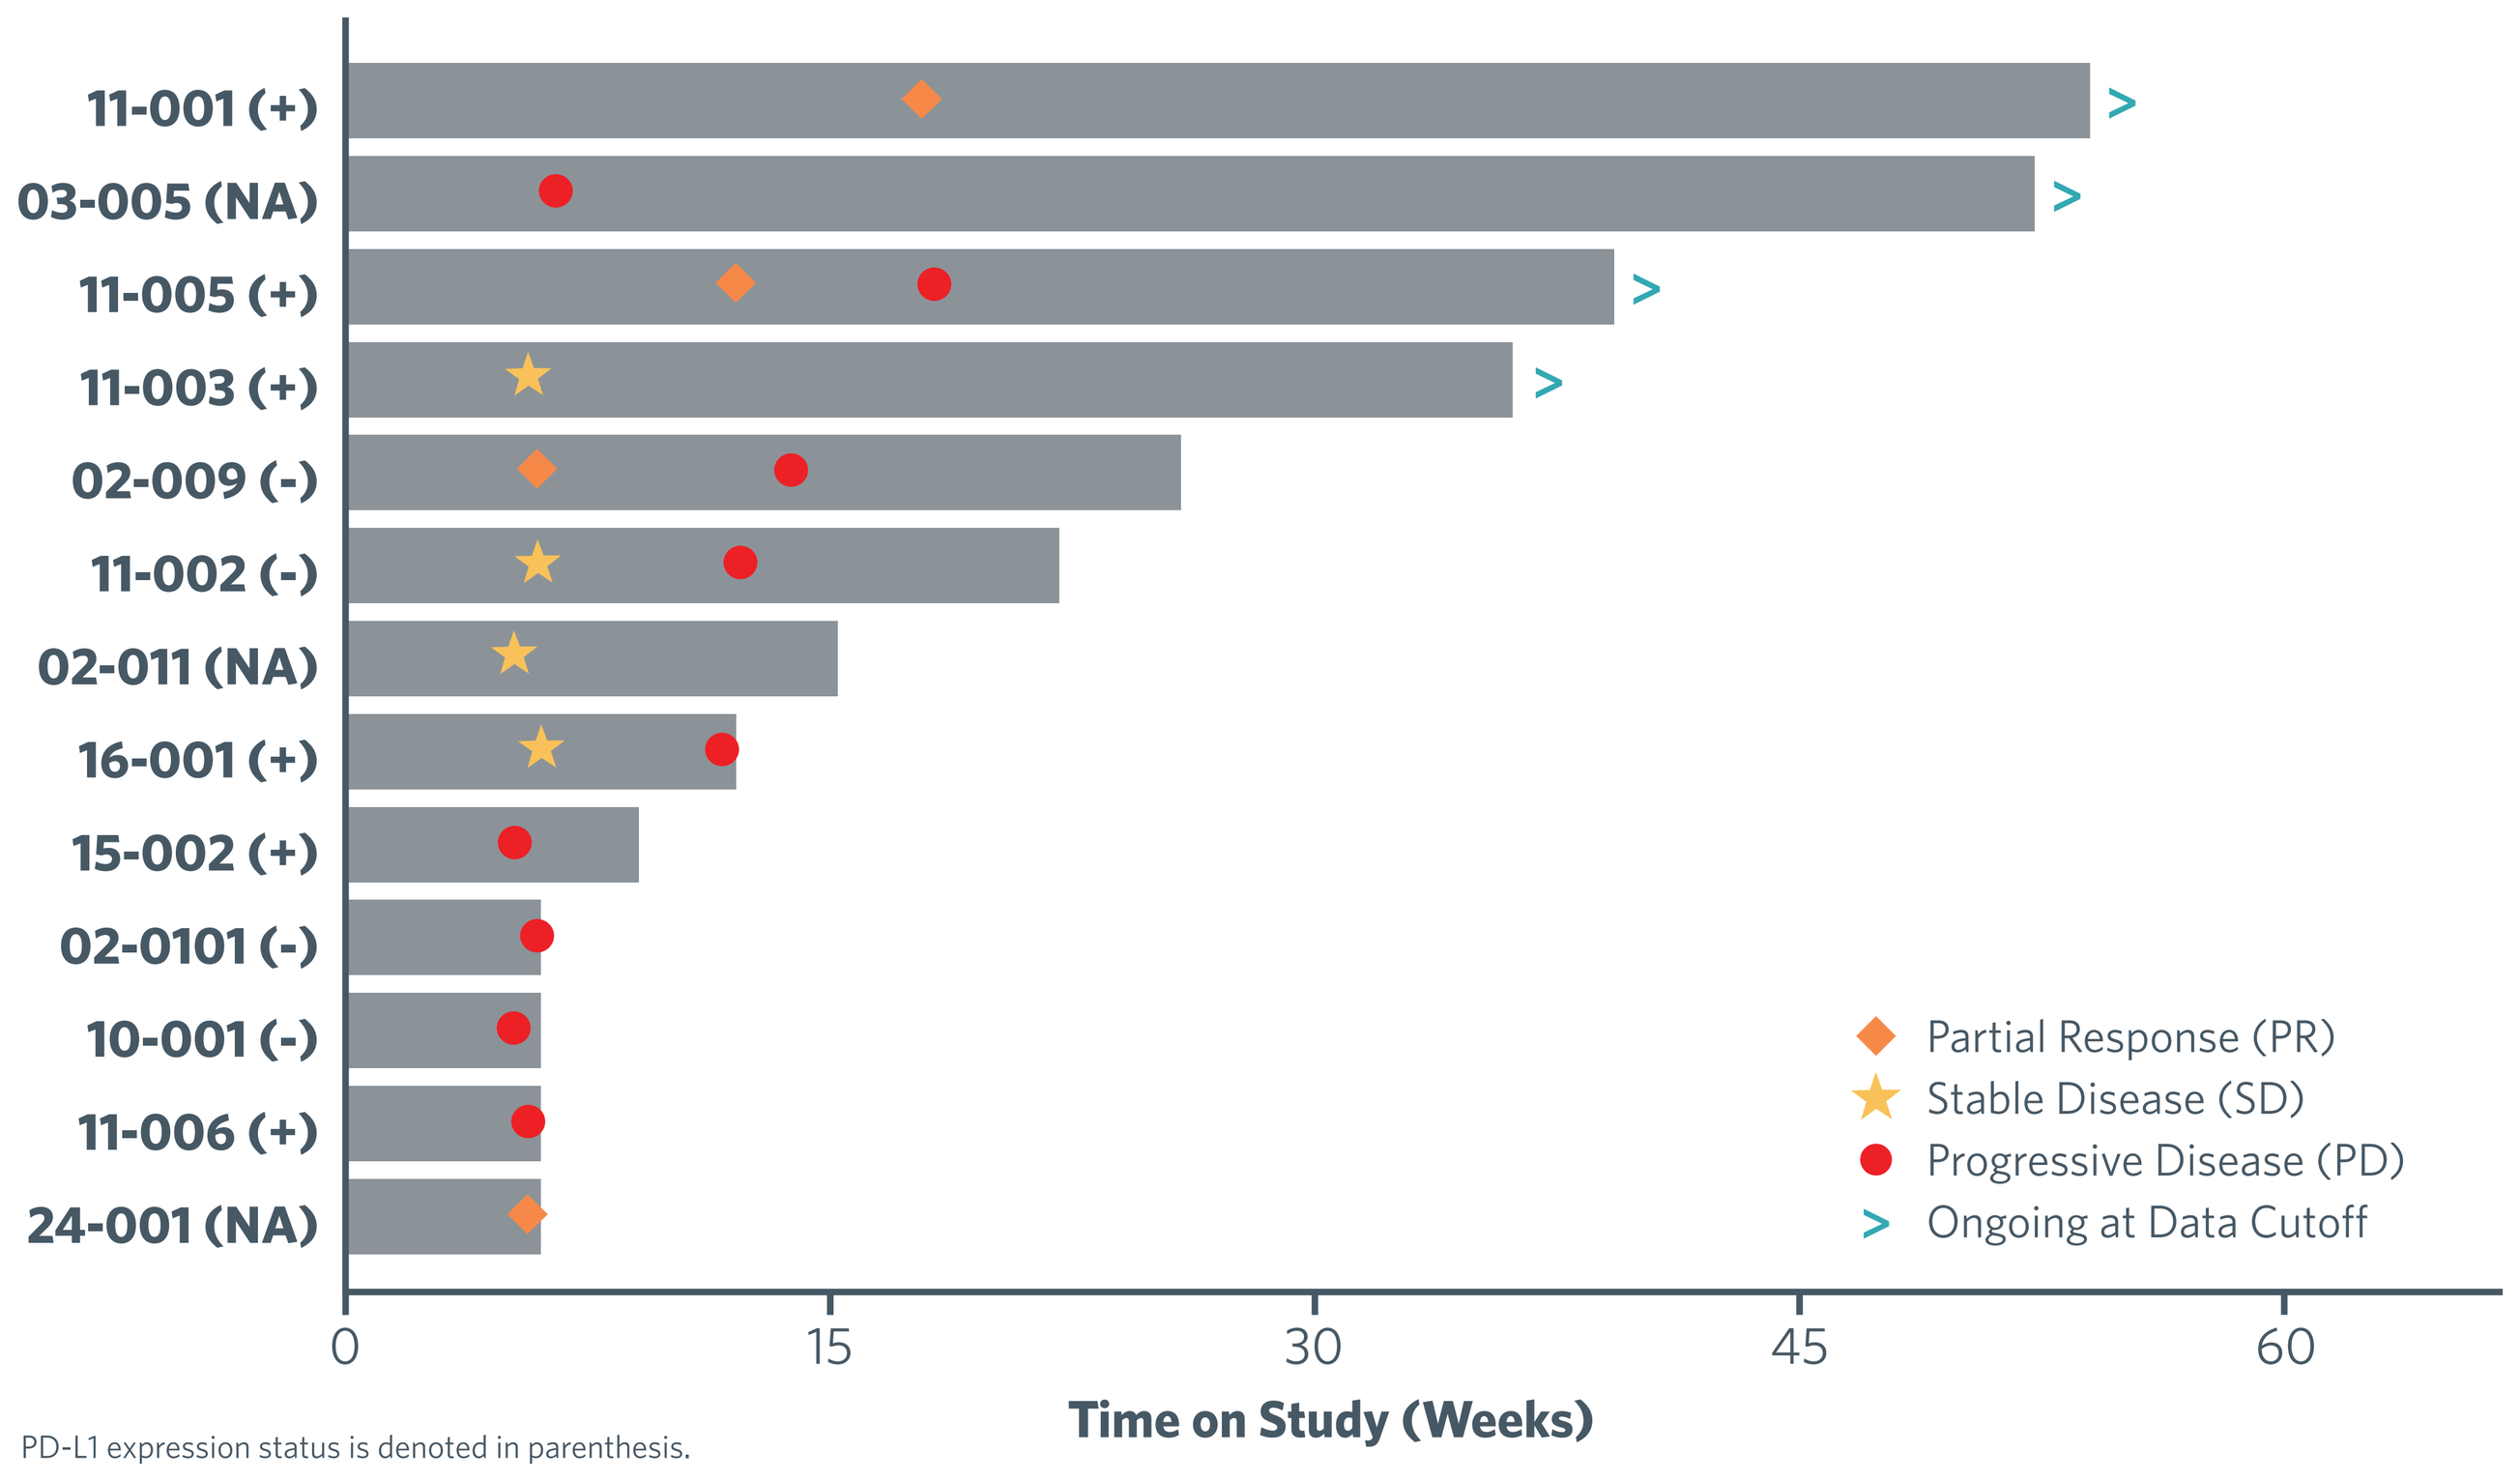
11-001 (+)
03-005 (NA)
11-005 (+)
11-003 (+)
02-009 (-)
11-002 (-)
02-011 (NA)
16-001 (+)
15-002 (+)
02-0101 (-)
10-001 (-)
11-006 (+)
24-001 (NA)
Partial Response (PR)
Stable Disease (SD)
Progressive Disease (PD)
Ongoing at Data Cutoff
0
15
30
45
60
Time on Study (Weeks)

PD-L1 expression status is denoted in parenthesis.

Safety Summary

- With an additional 4.5 months of follow-up, no additional safety concerns were identified
- 13 patients (100%) reported a treatment-emergent adverse event (TEAE); 8 (62%) experienced Grade ≥3 TEAEs, and, of these, 4 experienced Grade ≥3 TEAEs related to treatment with either study drug[3]
 — 1 (8%) patient discontinued because of a TEAE

Results of Exploratory Objectives

- Gene expression analysis of samples pre- and posttreatment suggest that tumor microenvironment becomes more inflamed after treatment with ENT + PEMBRO

Figure 3. **Tumor Inflammation Signature (TIS) Scores Were Analyzed Pre- and Posttreatment**

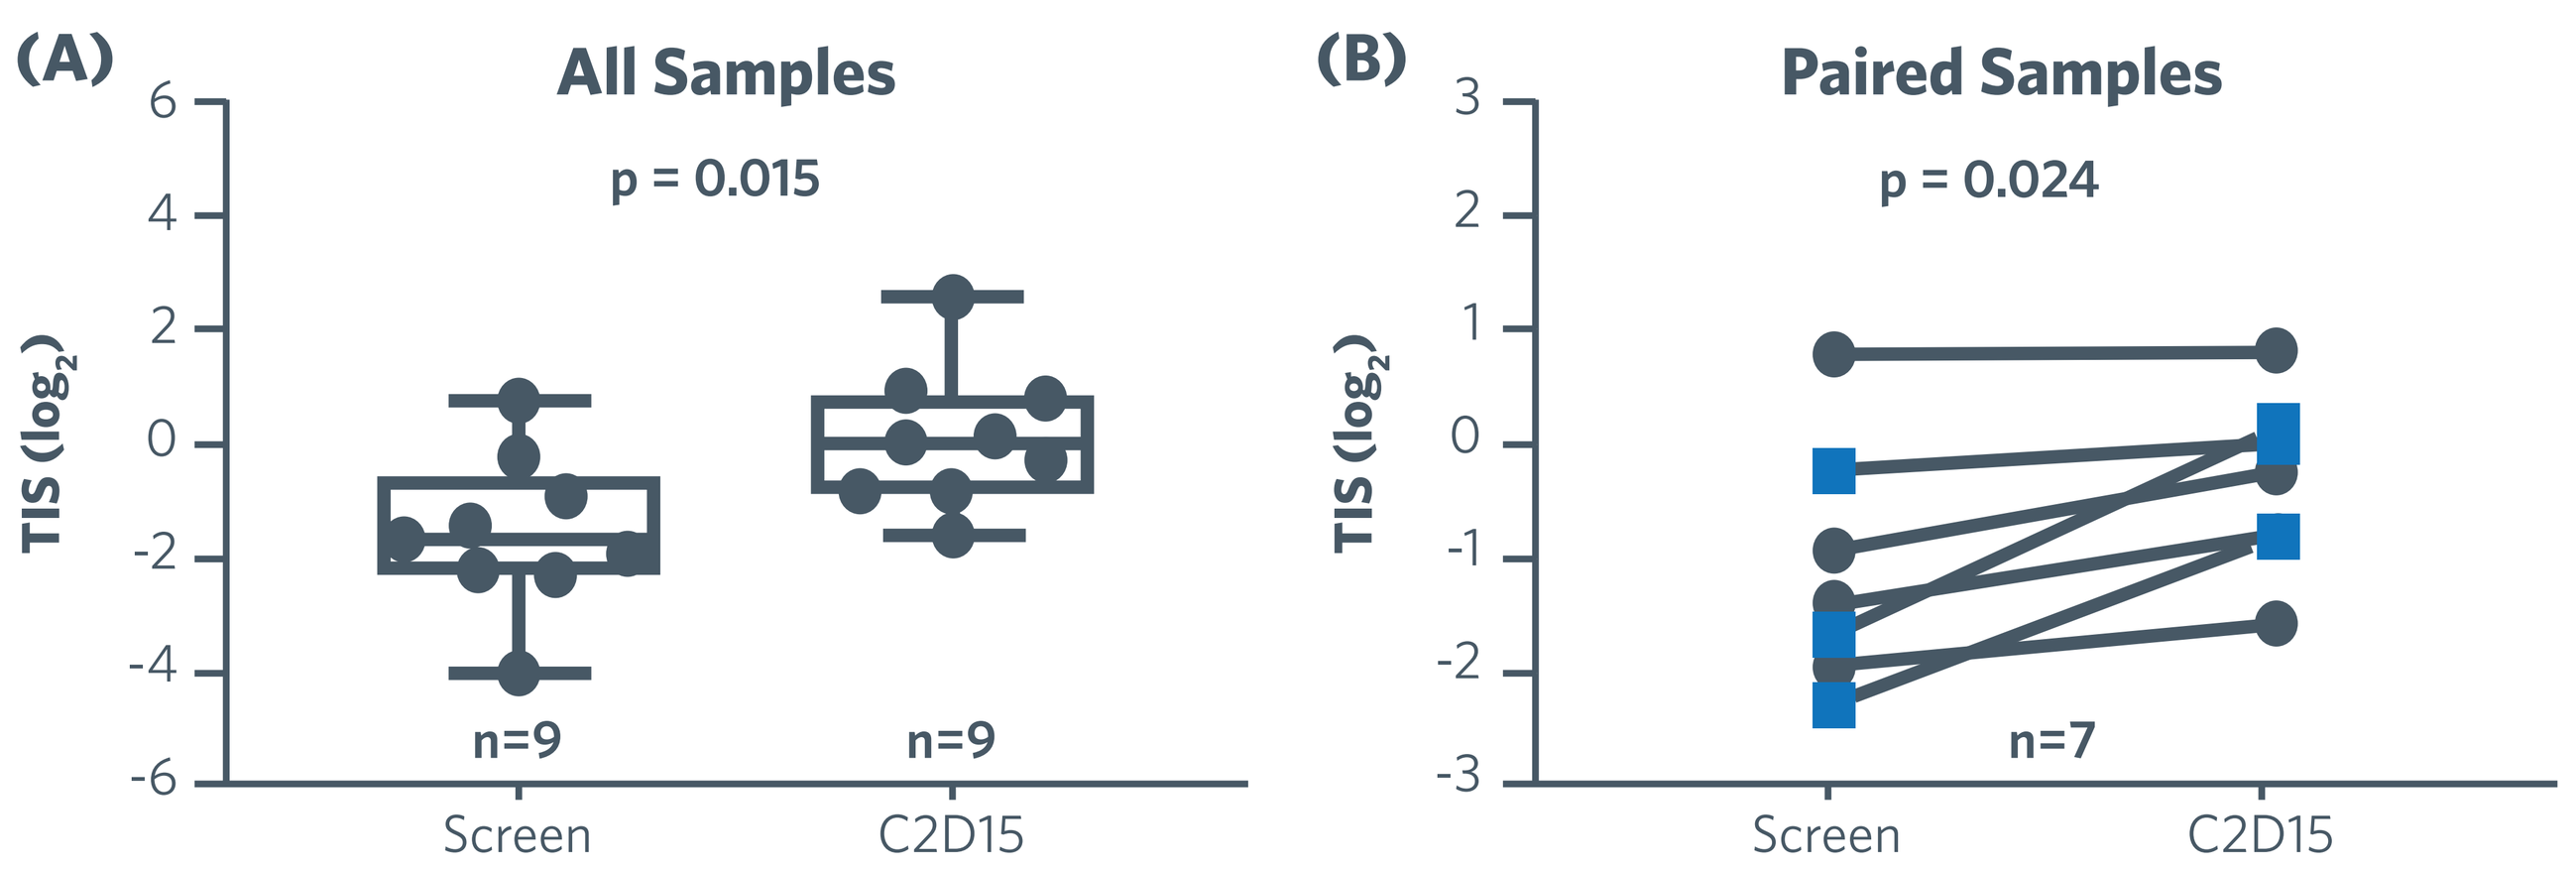
(A)
All Samples
p = 0.015
TIS (log_2)
n=9
n=9
Screen
C2D15
(B)
Paired Samples
p = 0.024
TIS (log_2)
n=7
Screen
C2D15

TIS = Tumor Inflammation Signature.

RESULTS *Continued*

- Significant increase in TIS observed when pooled pre- and postdose samples were compared (**Figure 3A**), suggesting elevated tumor microenvironment inflammation
- In a subset of patient matched samples, 2 of 3 responding patients showed a robust TIS Increase (**Figure 3B**)
- After treatment, the tumor microenvironment becomes more supportive of T-cell activity (**Figure 4A**)
- IO 360 signature scores for IFN-γ (**Figure 4B**), CD8 T-cells (**Figure 4C**), and Cytotoxic Cells (**Figure 4D**) were elevated, suggesting enhanced support after treatment for cytotoxic T-cell activity in the tumor microenvironment

Figure 4. **Heatmap Analysis Suggests That Gene Expression Changes Posttreatment were Associated with Enhanced Immune Function (A); IO 360 Signature Scores for IFN-γ (B), CD8 T-cells (C), and Cytotoxic Cells (D)**

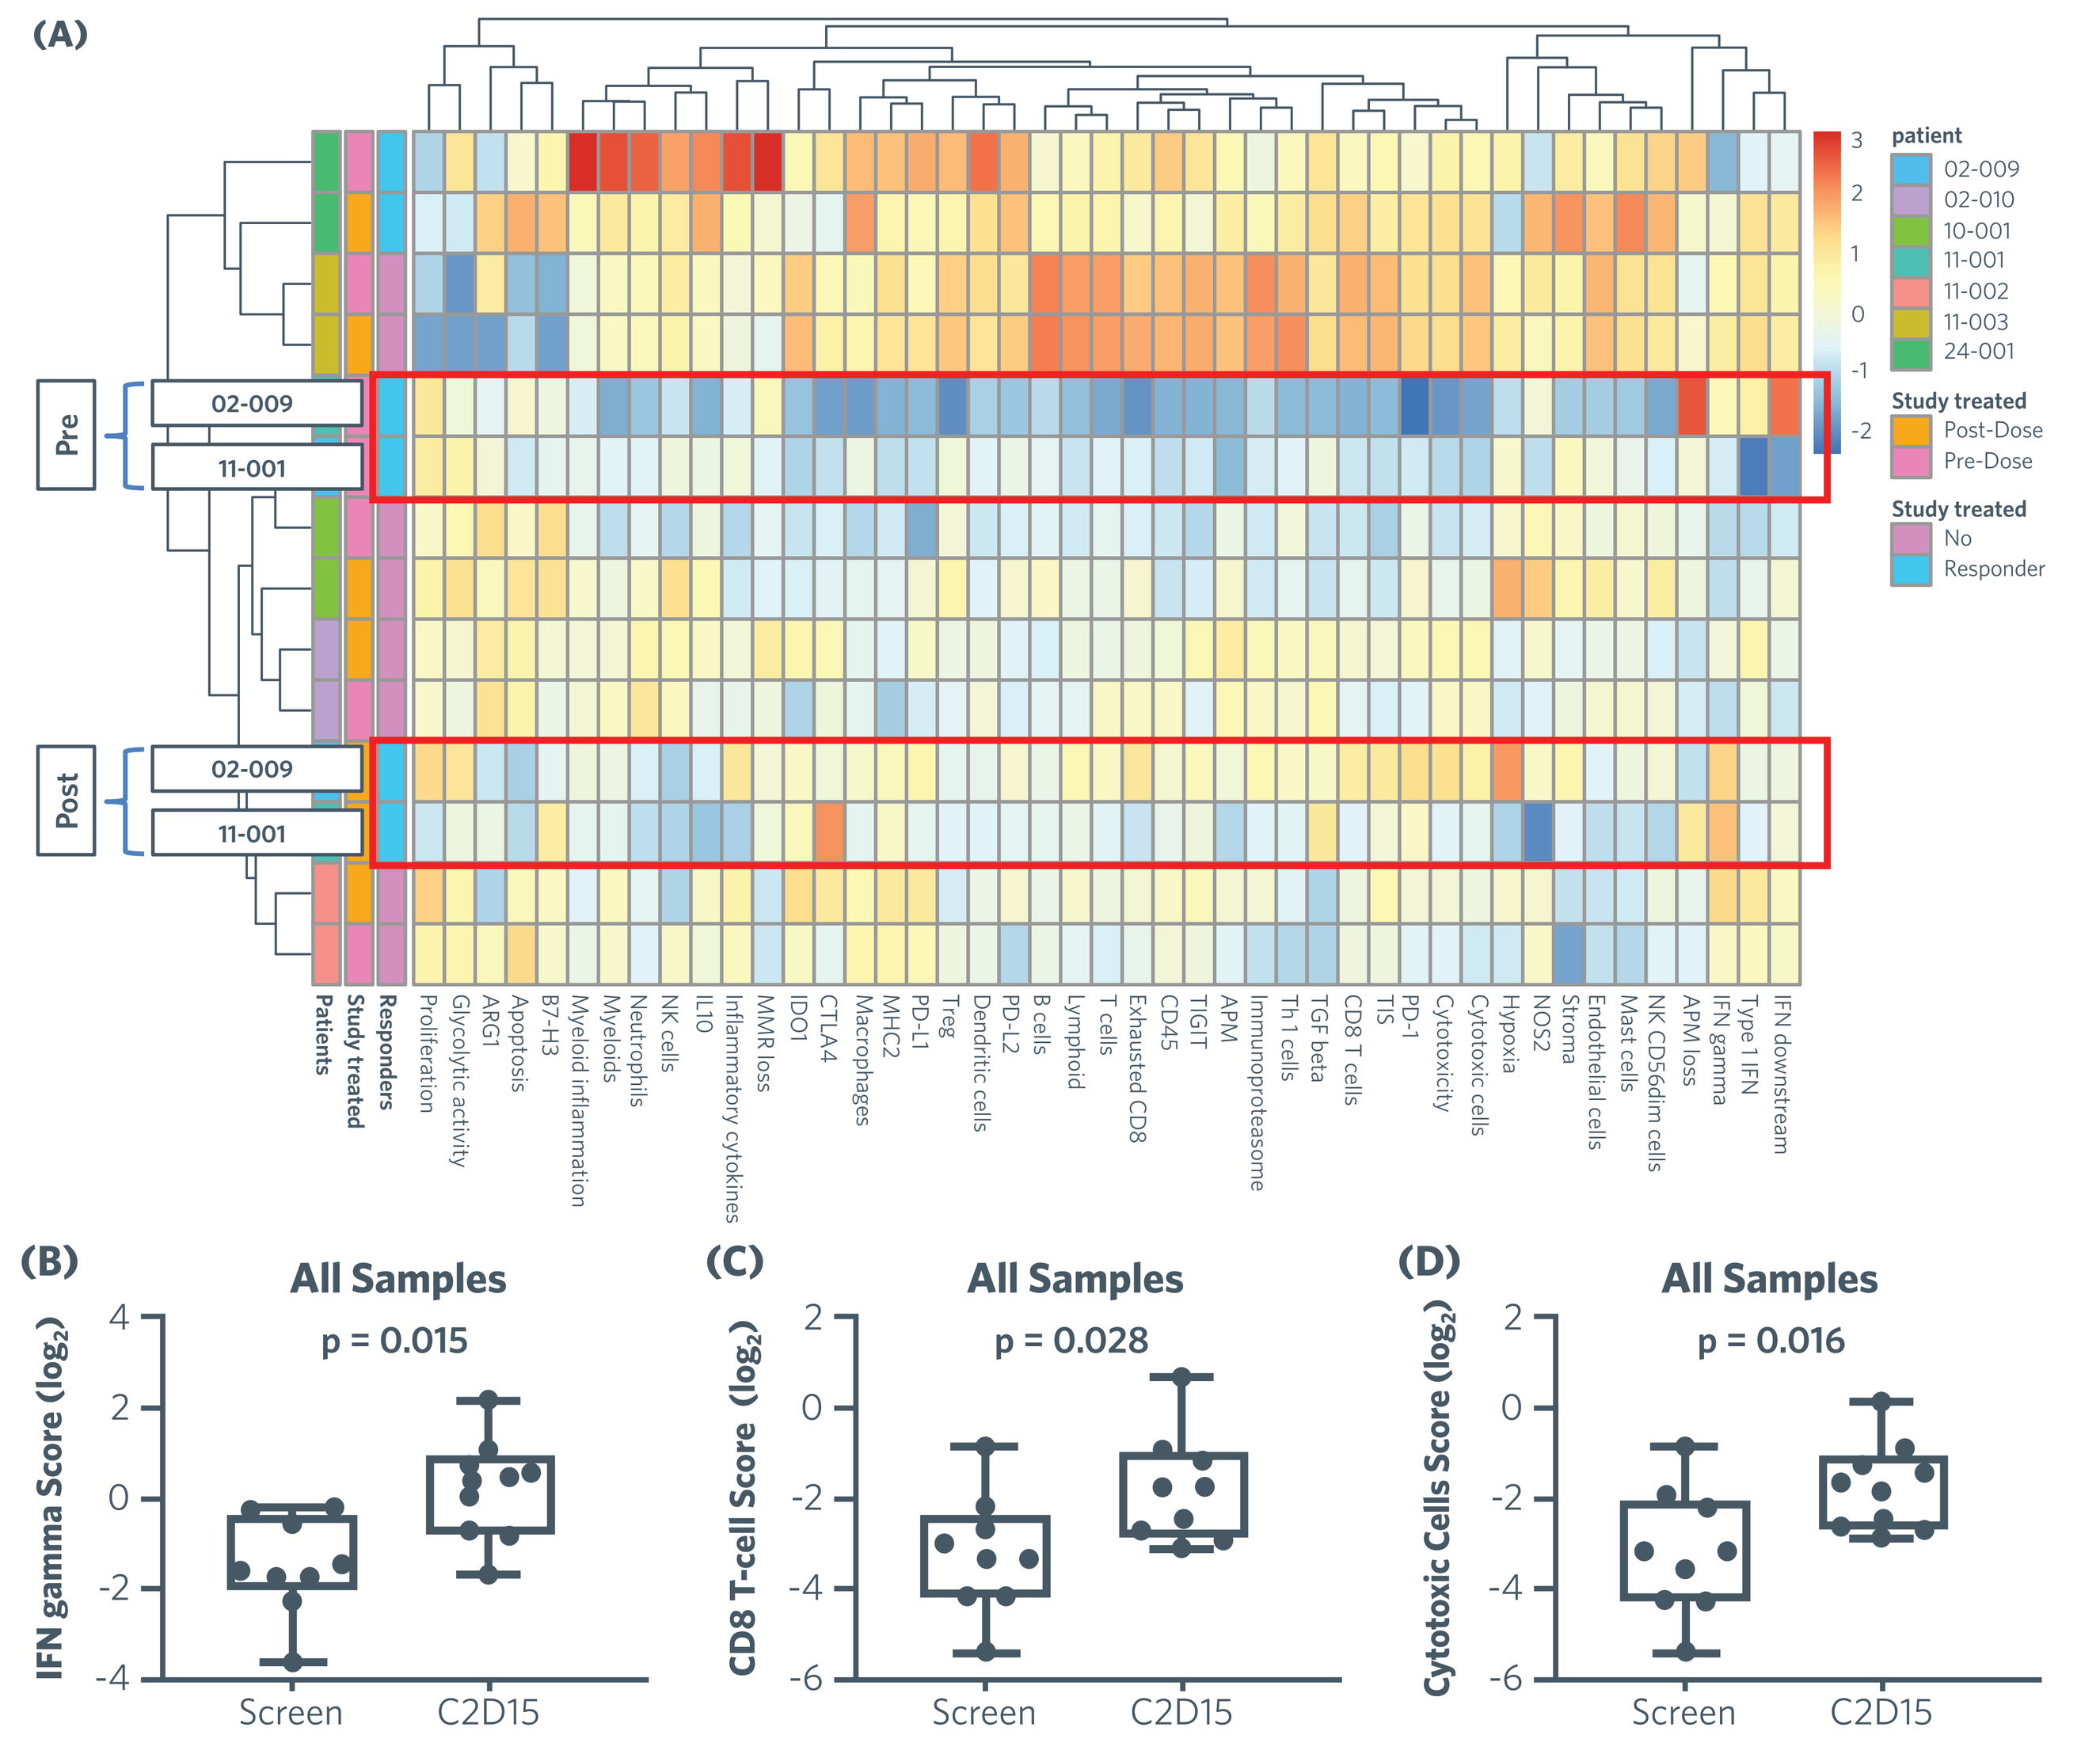
(A)
patient
02-009
02-010
10-001
11-001
11-002
11-003
24-001
Study treated
Post-Dose
Pre-Dose
Study treated
No
Responder
Pre
Post
02-009
11-001
Patients
Study treated
Responders
Proliferation
Glycolytic activity
ARG1
Apoptosis
B7-H3
Myeloid inflammation
Myeloids
Neutrophils
NK cells
IL10
Inflammatory cytokines
MMR loss
IDO1
CTLA4
Macrophages
MHC2
PD-L1
Treg
Dendritic cells
PD-L2
B cells
Lymphoid
T cells
Exhausted CD8
CD45
TIGIT
APM
Immunoproteasome
Th1 cells
TGF beta
CD8 T cells
TIS
PD-1
Cytotoxicity
Cytotoxic cells
Hypoxia
NOS2
Stroma
Endothelial cells
Mast cells
NK CD56dim cells
APM loss
IFN gamma
Type 1 IFN
IFN downstream
(B)
All Samples
p = 0.015
IFN gamma Score (log_2)
(C)
All Samples
p = 0.028
CD8 T-cell Score (log_2)
(D)
All Samples
p = 0.016
Cytotoxic Cells Score (log_2)
Screen
C2D15

iCOS-iCOSL = inducible costimulatory-iCOS ligand; IFN= interferon; IPA = ingenuity pathway analysis; TH = T helper.

Figure 5. **Immunofluorescence Analysis for MDSCs and CD8+ T cells Were Analyzed Pre- and Posttreatment**

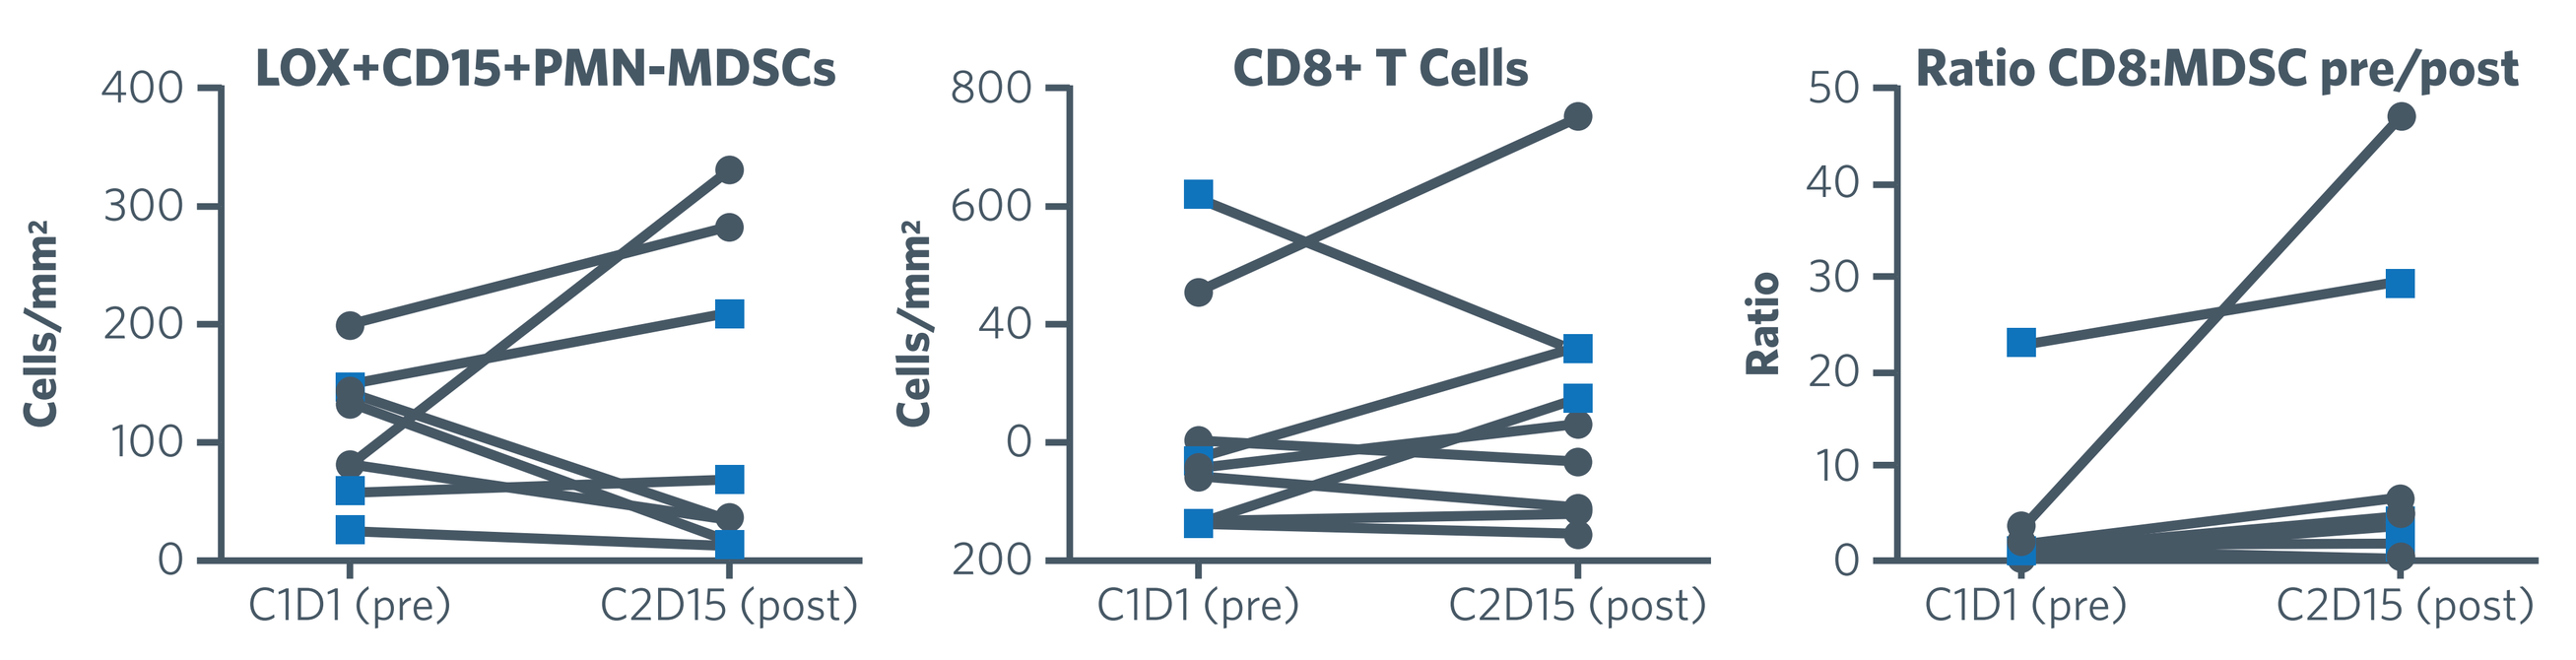
LOX+CD15+PMN-MDSCs
Cells/mm²
CD8+ T Cells
Cells/mm²
Ratio CD8:MDSC pre/post
Ratio
C1D1 (pre)
C2D15 (post)

MDSC = myeloid-derived suppressor cells; PBMC = peripheral blood mononuclear cell.

- Intratumor CD8+ T cell and MDSC changes are consistent with gene expression results and treatment effect as noted in **Figure 5**
 — CD8+ T cells and LOX1+CD15+ MDSCs measured by immunofluorescence
 — CD8:MDSCs ratio increased in 6 of 8 (75%) patients (Includes 3 PRs and 1 ongoing SD), which may suggest clinical benefit
 — Increases in both T cells and MDSCs were observed only in those tissue samples converted from noninflamed to inflamed
 — Overall, regardless of clinical outcome, a decrease in MDSCs (–35.7%; n=9) and an increase in CD8+ T cells (47.4%; n=9) between pre-and posttreatment biopsies were noted

SUMMARY AND CONCLUSIONS

- ENT in combination with PEMBRO exhibits clinical efficacy in melanoma patients previously exposed to immune checkpoint inhibition
- This combination has an acceptable toxicity profile
- Preliminary biomarker analysis supports the hypothesis that the addition of ENT restores inflammation in the tumor microenvironment necessary for successful re-treatment with an anti–PD-(L)1

References

1. Orillion A et al. *Clin Cancer Res.* 2017;23(17):5187-5201; **2.** Spranger S, Gajewski T. *J Immunother Cancer.* 2013;1:16; **3.** Johnson ML et al. *J Clin Oncol.* 2017;35(suppl):Abstr 9529; **4.** Simon R. *Oncology (Williston Park).* 1989;3(7):43-49; **5.** Ayers et al. *J Clin Invest.* 2017;127(8):2930-2940.

Acknowledgments

The authors thank the patients and their families, investigators and study staff, and NanoString® Technologies, Inc. This study was sponsored by Syndax Pharmaceuticals, Inc., in collaboration with Merck & Co., Inc., Kenilworth, NJ. Writing and editorial assistance was provided by Adelphi Communications, New York. This assistance was funded by Syndax Pharmaceuticals, Inc.

The QR code is intended to provide scientific information for individual reference. The PDF should not be altered or reproduced in any way.

Presented at the Society for the Immunotherapy of Cancer (SITC) Annual Meeting; November 8–12, 2017; National Harbor, MD, USA